Combination Therapy of exoSTING™ and exoIL-12™ Activates Systemic Anti-Tumor Immunity

Katherine Kirwin, Su Chul Jang, Chang Ling Sia, Shil Patel, Kevin Dooley, Tong Zi, Kelvin Zhang, Yanyan Liu, Kyriakos D. Economides, Sriram Sathyanarayanan

Codiak BioSciences, Cambridge, MA

SITC 2021 Washington, D.C.

Poster #572

Introduction

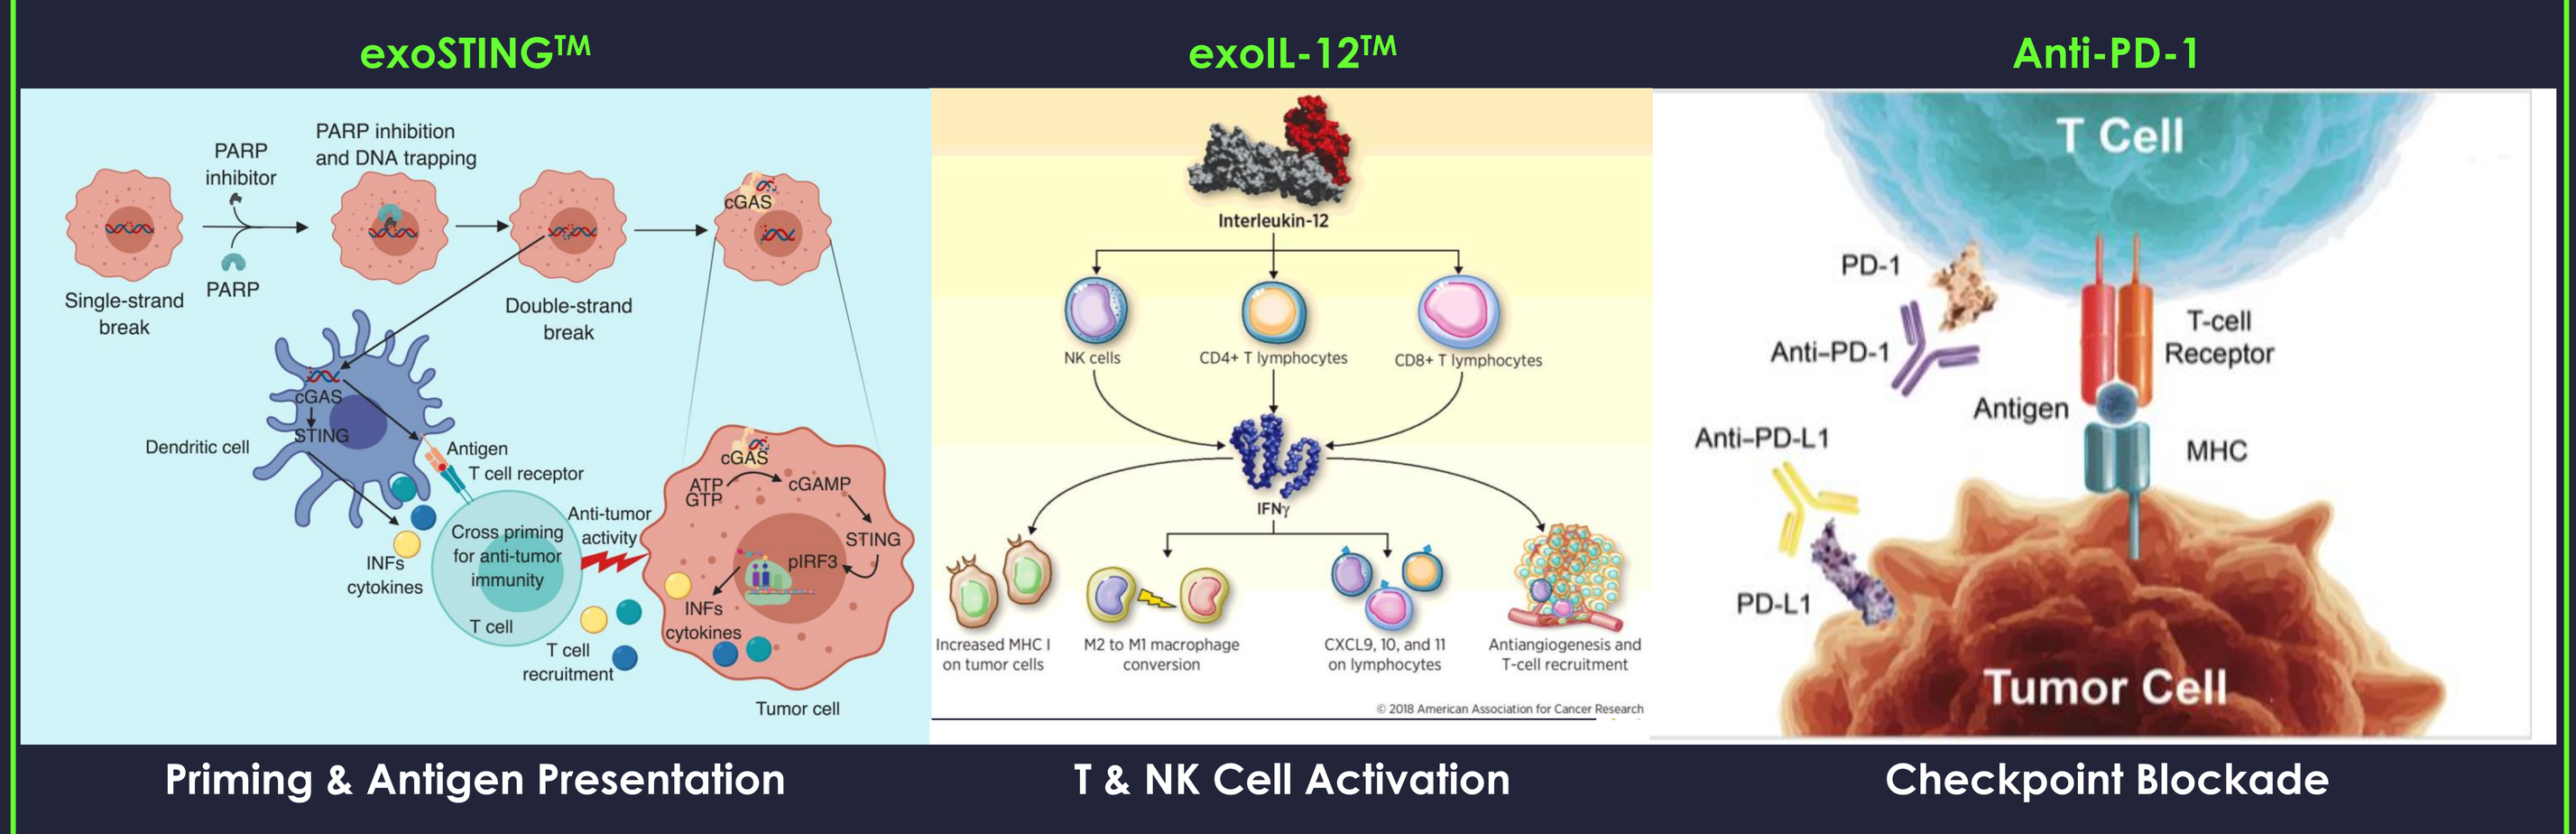
exoSTING™
exoIL-12™
Anti-PD-1
T Cell
PD-1
Anti-PD-1
T-cell Receptor
Antigen
Anti-PD-L1
MHC
PD-L1
Tumor Cell
Priming & Antigen Presentation
T & NK Cell Activation
Checkpoint Blockade

- Exosomes are natural, cell-derived vesicles that act as a non-immunogenic delivery system for multiple therapeutic payloads.
- exoSTING™, composed of engineered exosomes overexpressing PTGFRN and loaded with a cyclic dinucleotide (CDN) small molecule STING agonist, leverages the inherent ability of exosomes to deliver STING agonists selectively to antigen presenting cells (APCs). exoSTING selectively activates the STING pathway in tumor-resident APCs, demonstrating greater potency than free STING agonist (FSA) and without immune ablation.
- exoIL-12™ is composed of exosomes engineered to overexpress functional IL-12 attached via fusion to PTGFRN. exoIL-12 limits systemic exposure and maximizes local pharmacology within the tumor microenvironment.
- Here we show that the combination of exoSTING and exoIL-12 with the addition of anti-PD-1 maximizes systemic anti-tumor immunity.

References:
- Jang, S.C., Economides, K.D., Moniz, R.J. *et al.* ExoSTING, an extracellular vesicle loaded with STING agonists, promotes tumor immune surveillance. *Commun Biol* **4**, 497 (2021).
- Lewis, N.D., Sia, C.L., Kirwin, K. *et al*. Exosome Surface Display of IL12 Results in Tumor-Retained Pharmacology with Superior Potency and Limited Systemic Exposure Compared with Recombinant IL12. *Molecular Cancer Therapeutics* **20**, 3, pp. 523–534 (2020).
- Berraondo, P., *et al*. Revisiting interleukin-12 as a cancer immunotherapy agent. *Clin Cancer Res* **24**, 12, pp. 2716-2718 (2018).
- Drake, C.G., *et al.* Breathing new life into immunotherapy: review of melanoma, lung and kidney cancer. *Nat Rev Clin Oncol.* 11, 1, pp. 24-37 (2014).

1. Potent single agent anti-tumor immunity by exoSTING and exoIL-12 in mouse tumor models

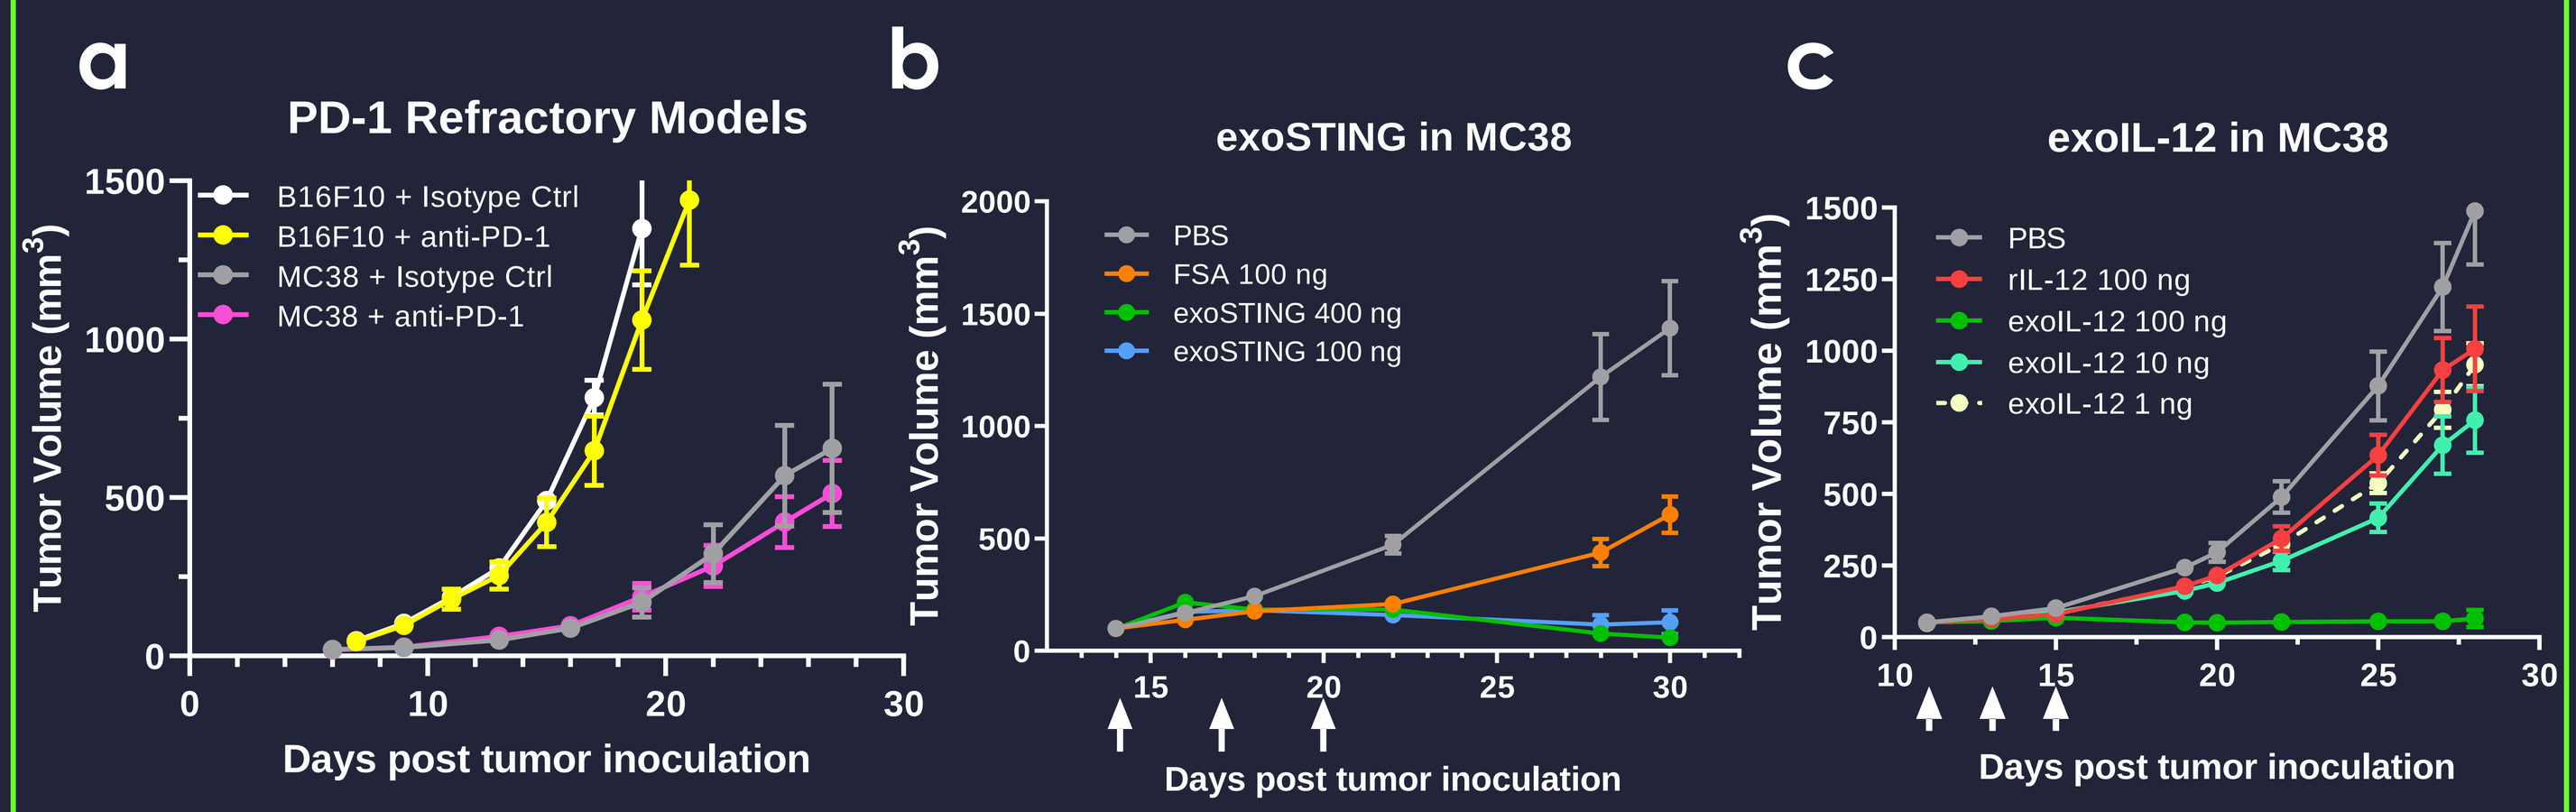
a
PD-1 Refractory Models
B16F10 + Isotype Ctrl
B16F10 + anti-PD-1
MC38 + Isotype Ctrl
MC38 + anti-PD-1
Tumor Volume (mm³)
Days post tumor inoculation
b
exoSTING in MC38
PBS
FSA 100 ng
exoSTING 400 ng
exoSTING 100 ng
c
exoIL-12 in MC38
PBS
rIL-12 100 ng
exoIL-12 100 ng
exoIL-12 10 ng
exoIL-12 1 ng

(a) Subcutaneous MC38 (n = 10) or B16F10 (n = 5) tumors are anti-PD-1 refractory. Monotherapy involving three times intratumoral (IT) dosing of (b) exoSTING and (c) exoIL12 dose-dependently reduced mean tumor volume in a MC38 subcutaneous mouse model compared to FSA and rIL12 respectively (n = 5). Arrows indicate days of intratumoral dosing.

2. exoSTING monotherapy reduces tumor FDG signal in non-injected mouse tumors

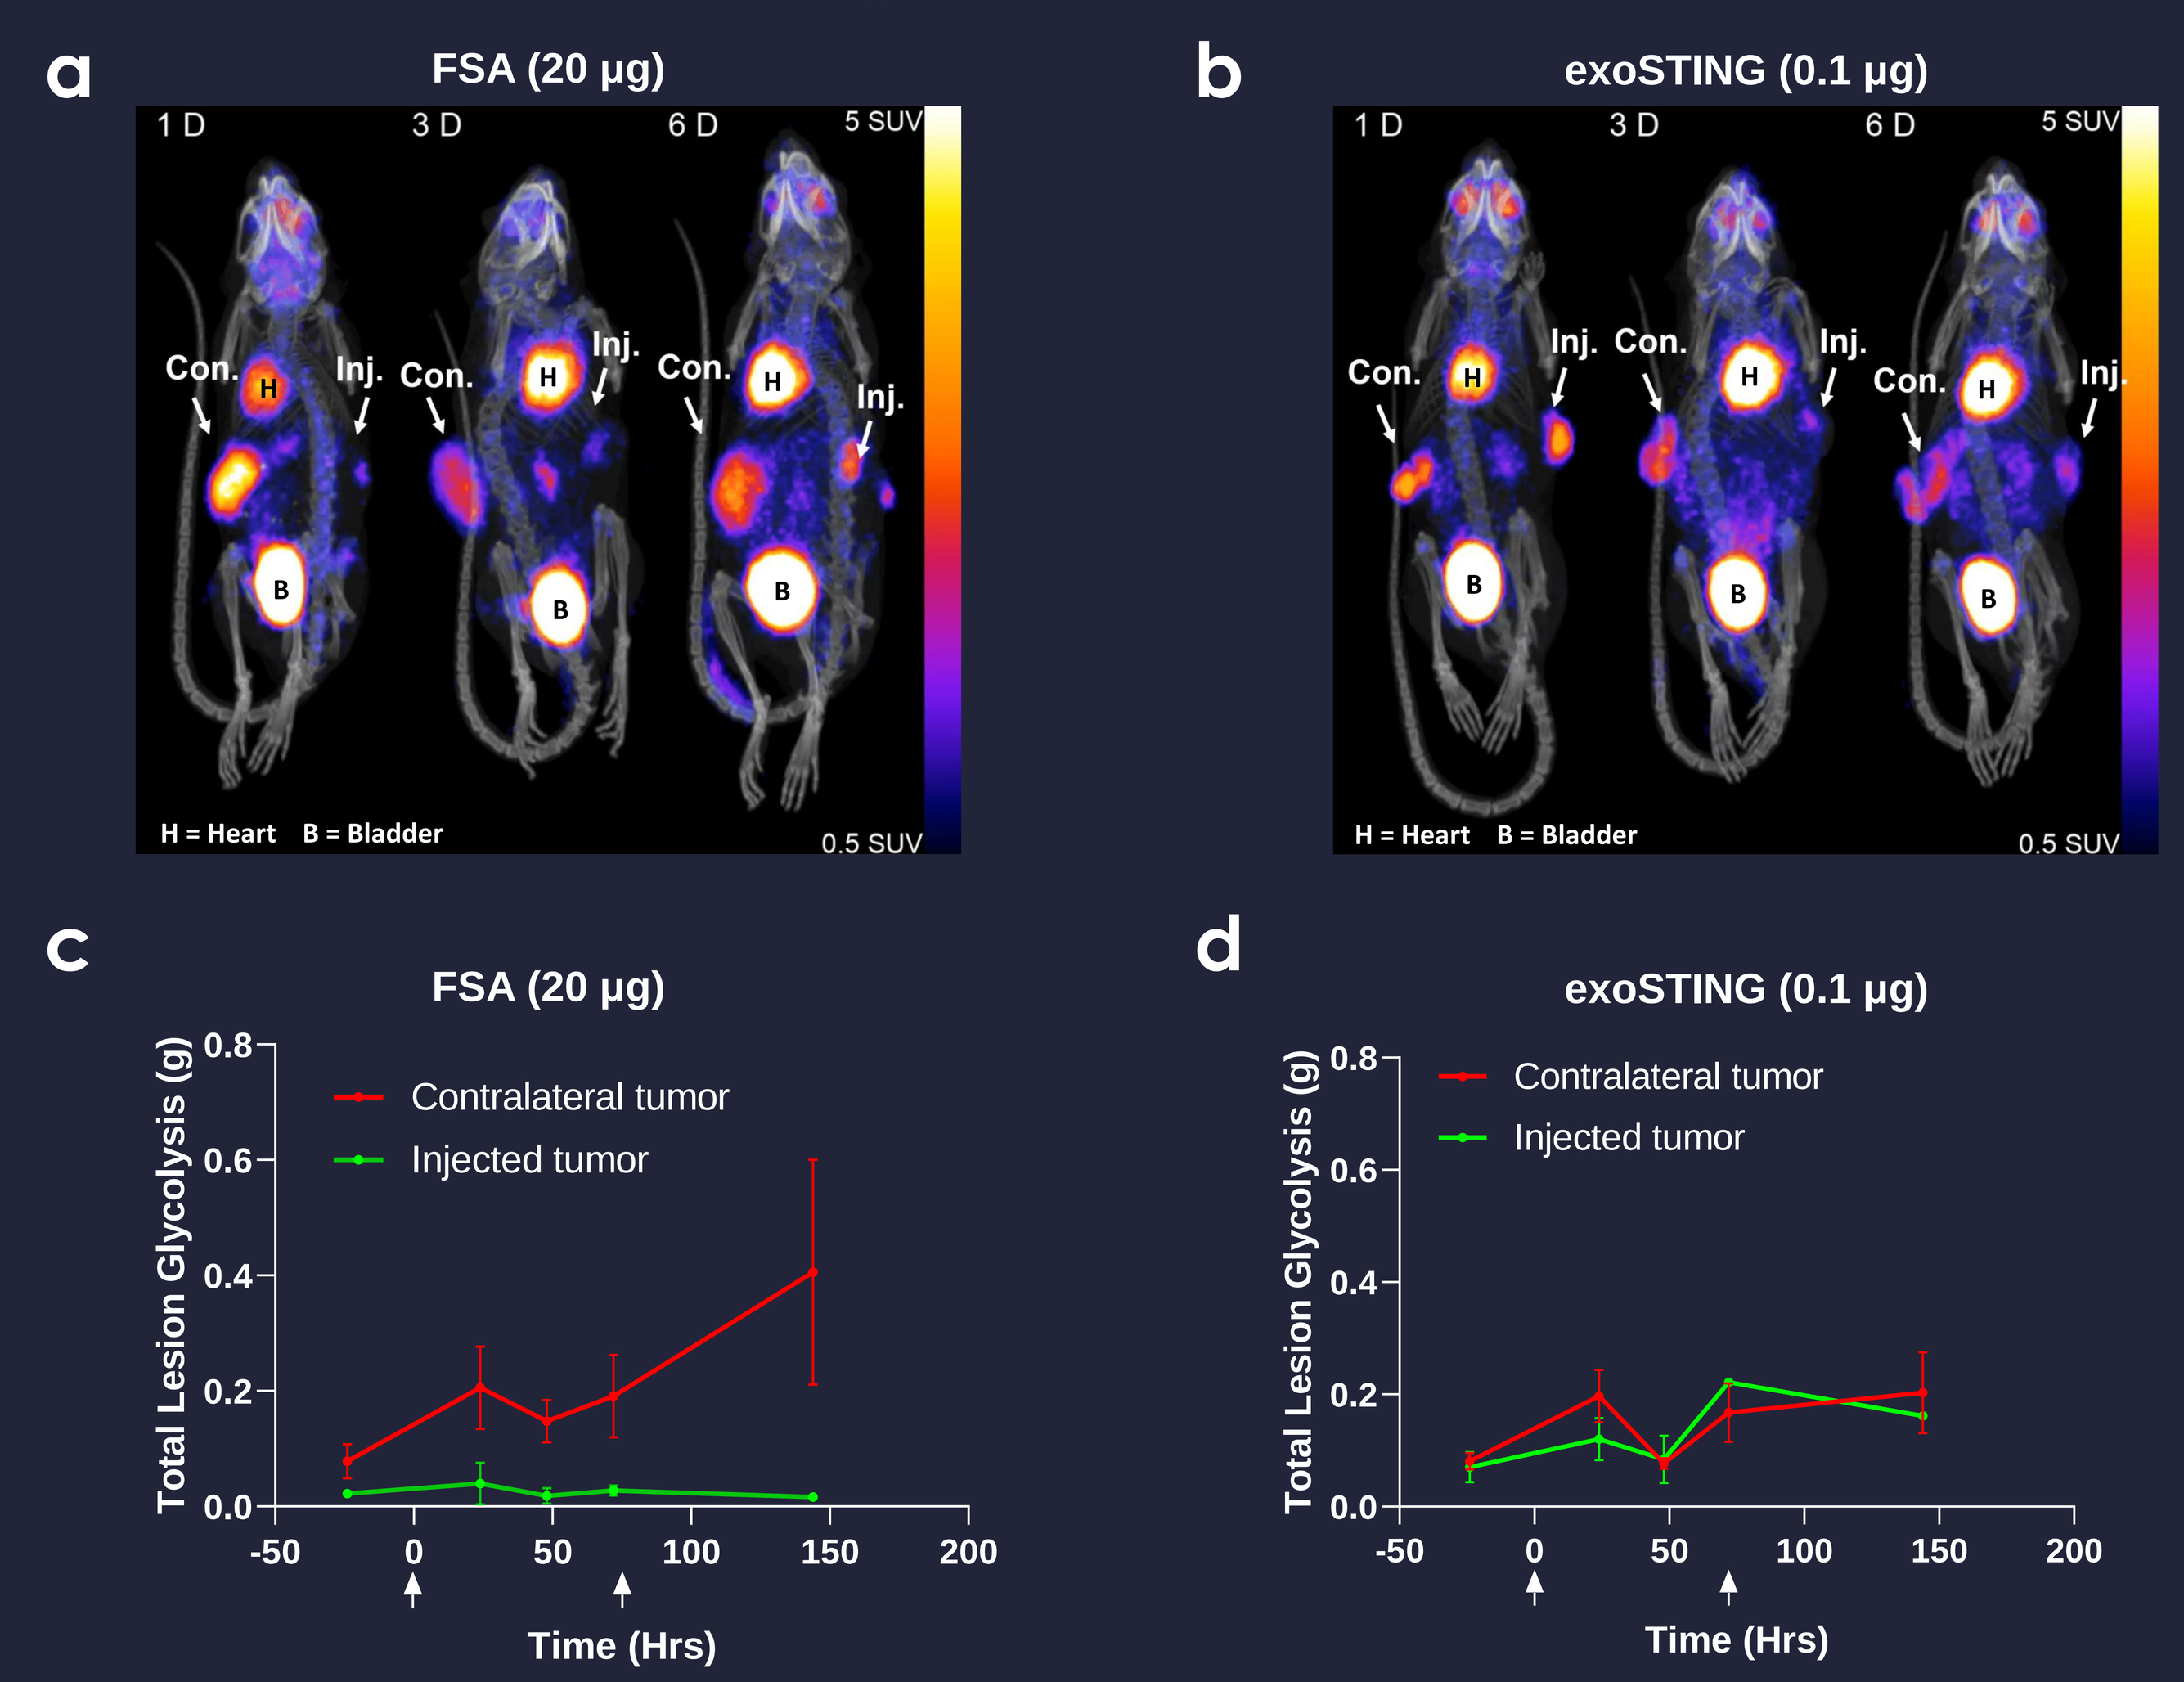
a
FSA (20 µg)
1 D
3 D
6 D
5 SUV
Con.
Inj.
H = Heart B = Bladder
0.5 SUV
b
exoSTING (0.1 µg)
c
FSA (20 µg)
Contralateral tumor
Injected tumor
Total Lesion Glycolysis (g)
Time (Hrs)
d
exoSTING (0.1 µg)

Representative PET-CT images of intratumoral free STING agonist (FSA, 20 µg) (a), or exoSTING (0.1 µg) (b), dosed into the right flank tumor 3 times a day at 3-day intervals in a subcutaneous dual flank B16F10 mouse model (n = 3-8 per group) and indicated by arrows in panels (c) and (d). [^{18}F]-FDG was injected intravenously at days -1, 1, 2, 3, and 6 with 10-minute static scans acquired an hour post-injection. Total lesion glycolysis was measured in both injected and non-injected contralateral tumors after FSA (20 µg) (c) and exoSTING (0.1 µg) (d) injection. Cont.; contralateral tumor, Inj.; injected tumor.

3. exoSTING, exoIL-12 combination reduces the growth of non-injected tumors

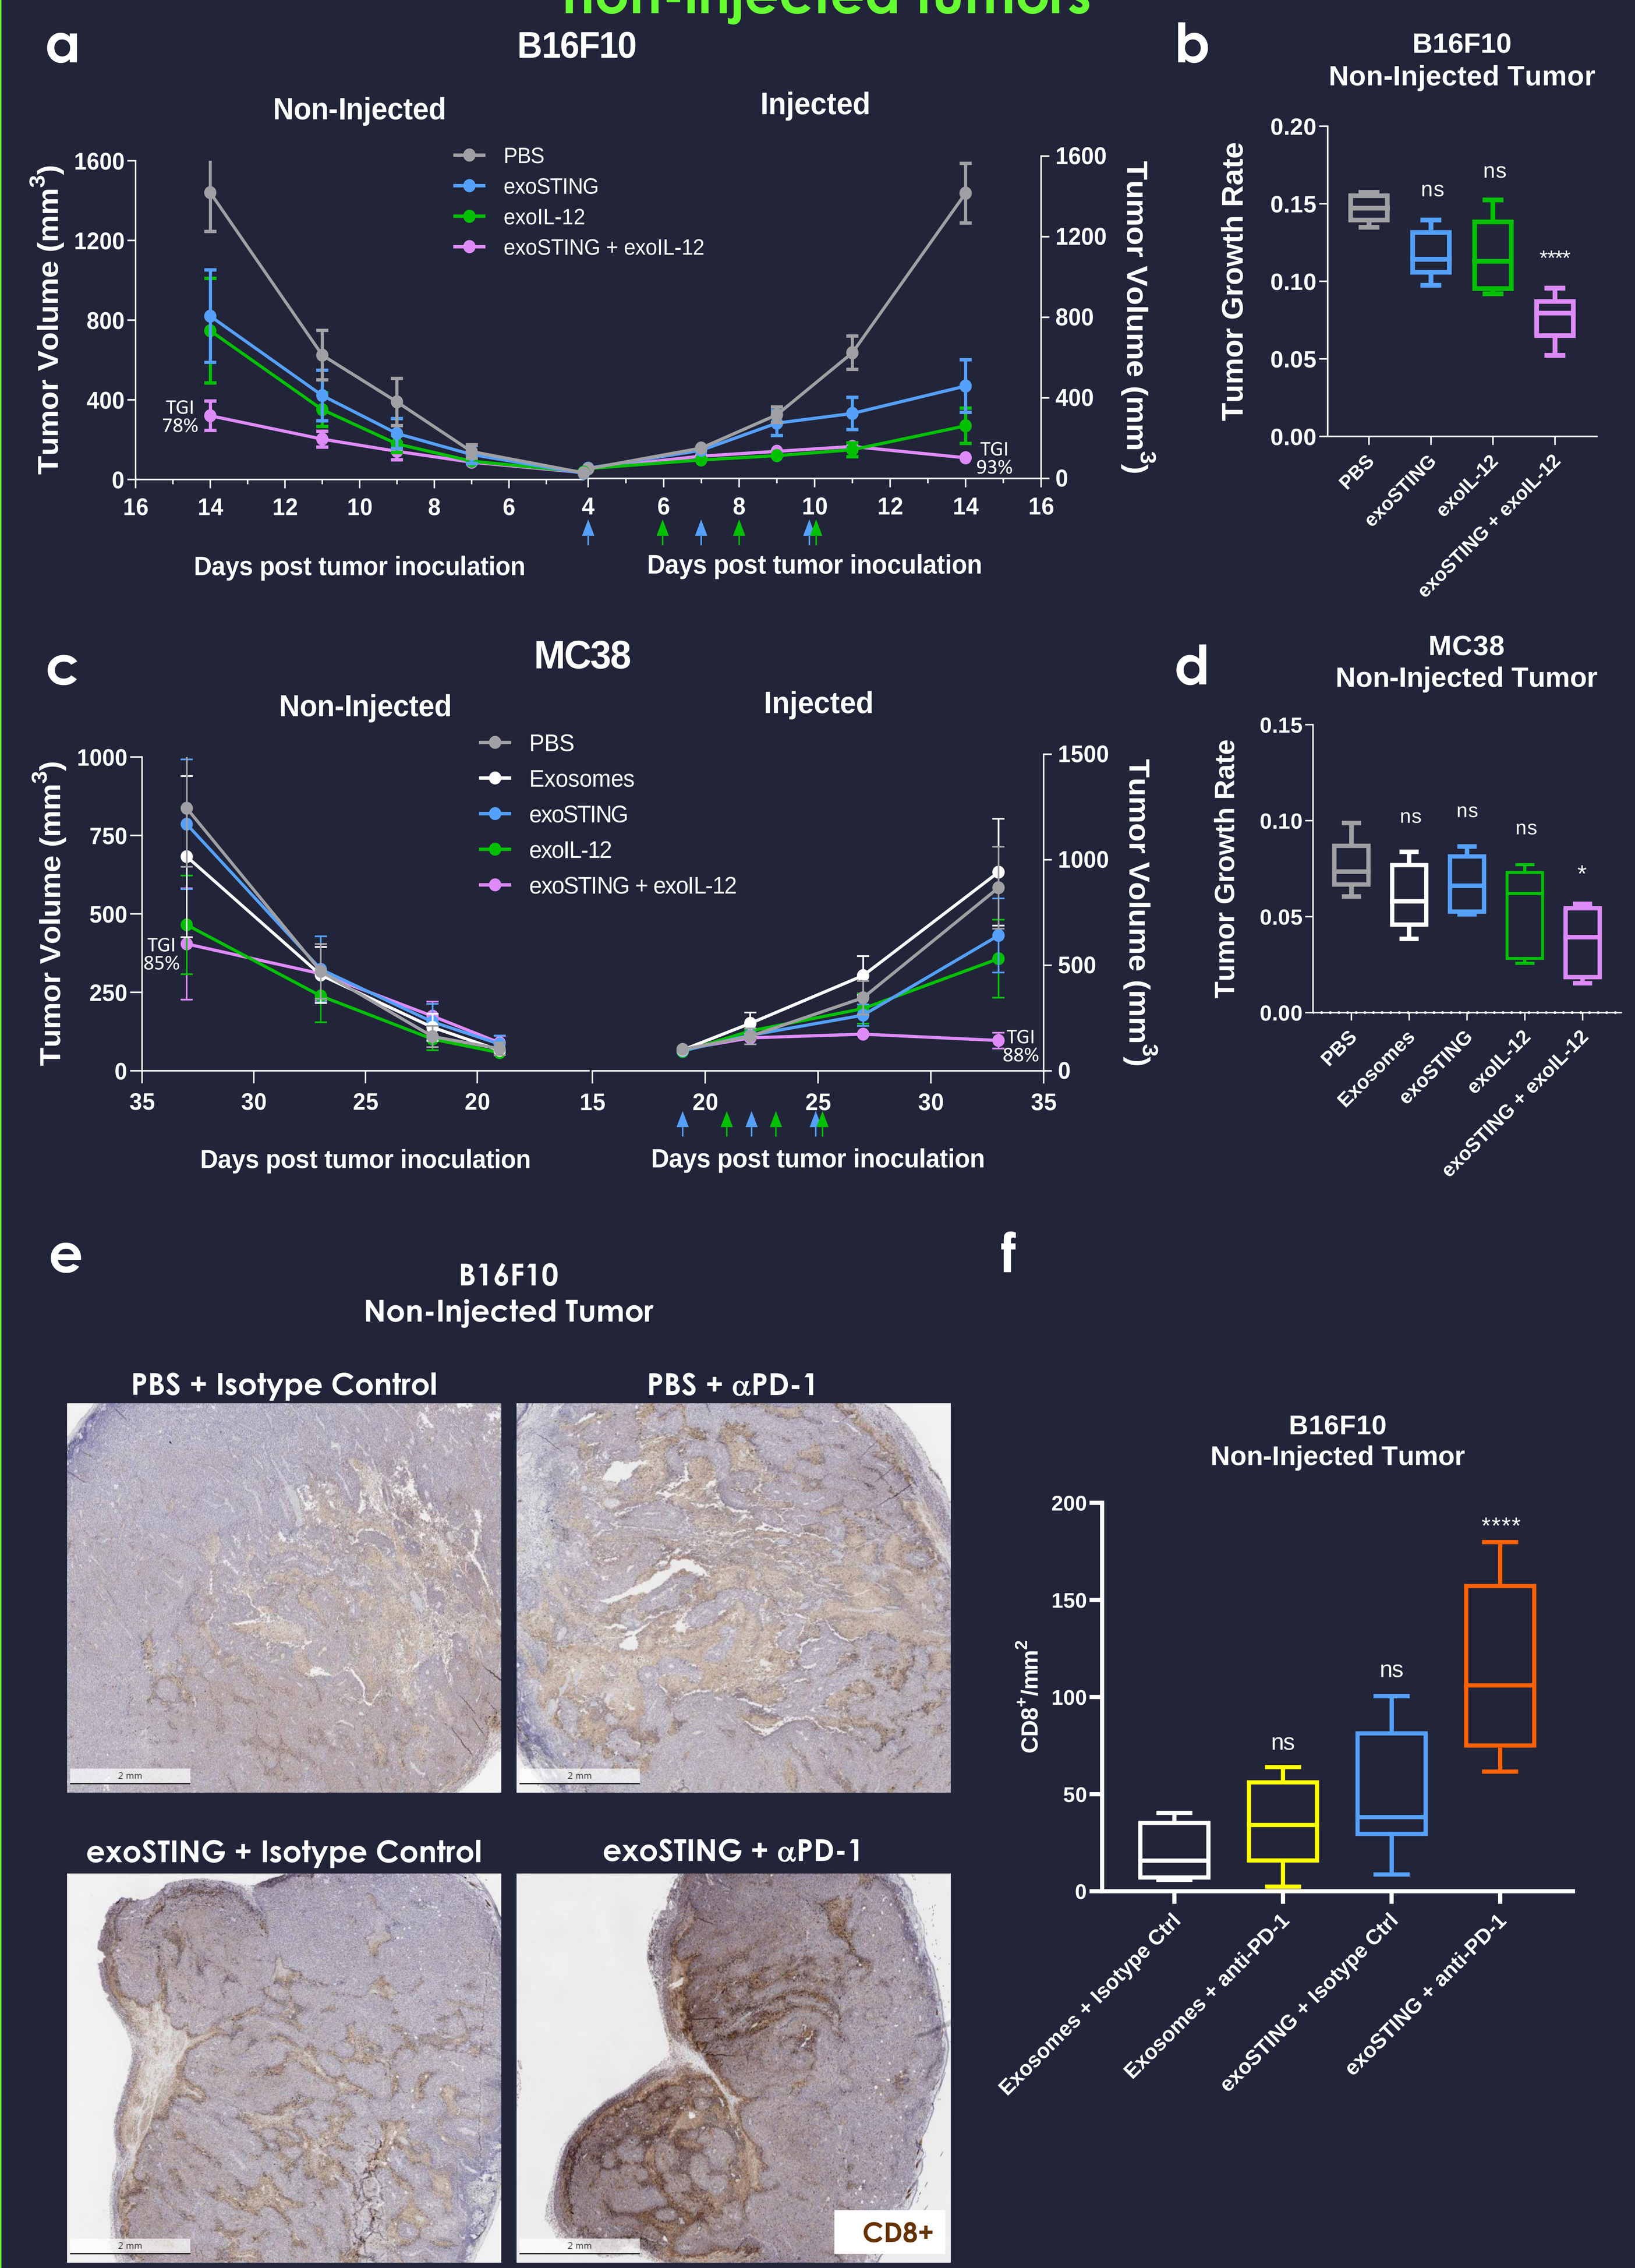
a
B16F10
Non-Injected
Injected
PBS
exoSTING
exoIL-12
exoSTING + exoIL-12
TGI 78%
TGI 93%
Tumor Volume (mm³)
Days post tumor inoculation
b
B16F10
Non-Injected Tumor
Tumor Growth Rate
c
MC38
Exosomes
TGI 85%
TGI 88%
d
MC38
Non-Injected Tumor
e
B16F10
Non-Injected Tumor
PBS + Isotype Control
PBS + αPD-1
exoSTING + Isotype Control
exoSTING + αPD-1
CD8+
f
B16F10
Non-Injected Tumor
CD8⁺/mm²
Exosomes + Isotype Ctrl
Exosomes + anti-PD-1
exoSTING + Isotype Ctrl
exoSTING + anti-PD-1

Dual combination therapy dosing of exoSTING (q3d) or exoIL12 (q2d) reduced mean tumor volume in a dual flank B16F10 (a) and MC38 (c) subcutaneous mouse model (n = 5). Dosing scheme indicated by arrows below the X-axis. Tumor growth rate of non-injected tumor (b,d) showed significant differences between PBS and exoSTING + exoIL-12 (one-way ANOVA with Tukey's multiple comparison test; **** $p<0.0001$, * $p=0.0230$, ns = Non-Significant). Non-injected B16F10 tumors were stained for CD8⁺ cells by immuno-histochemistry (shown in brown). Cross sections (e) and cumulative analysis (f) of CD8⁺ cells/mm² demonstrate increased CD8⁺ T cell infiltration when mice are dosed with exoSTING intratumorally and anti-PD-1 intraperitoneally (one-way ANOVA with Tukey's multiple comparison test; **** $p<0.0001$, ns = Non-Significant).

4. exoSTING, exoIL-12, anti-PD-1 triple combination eliminates both injected and non-injected tumors

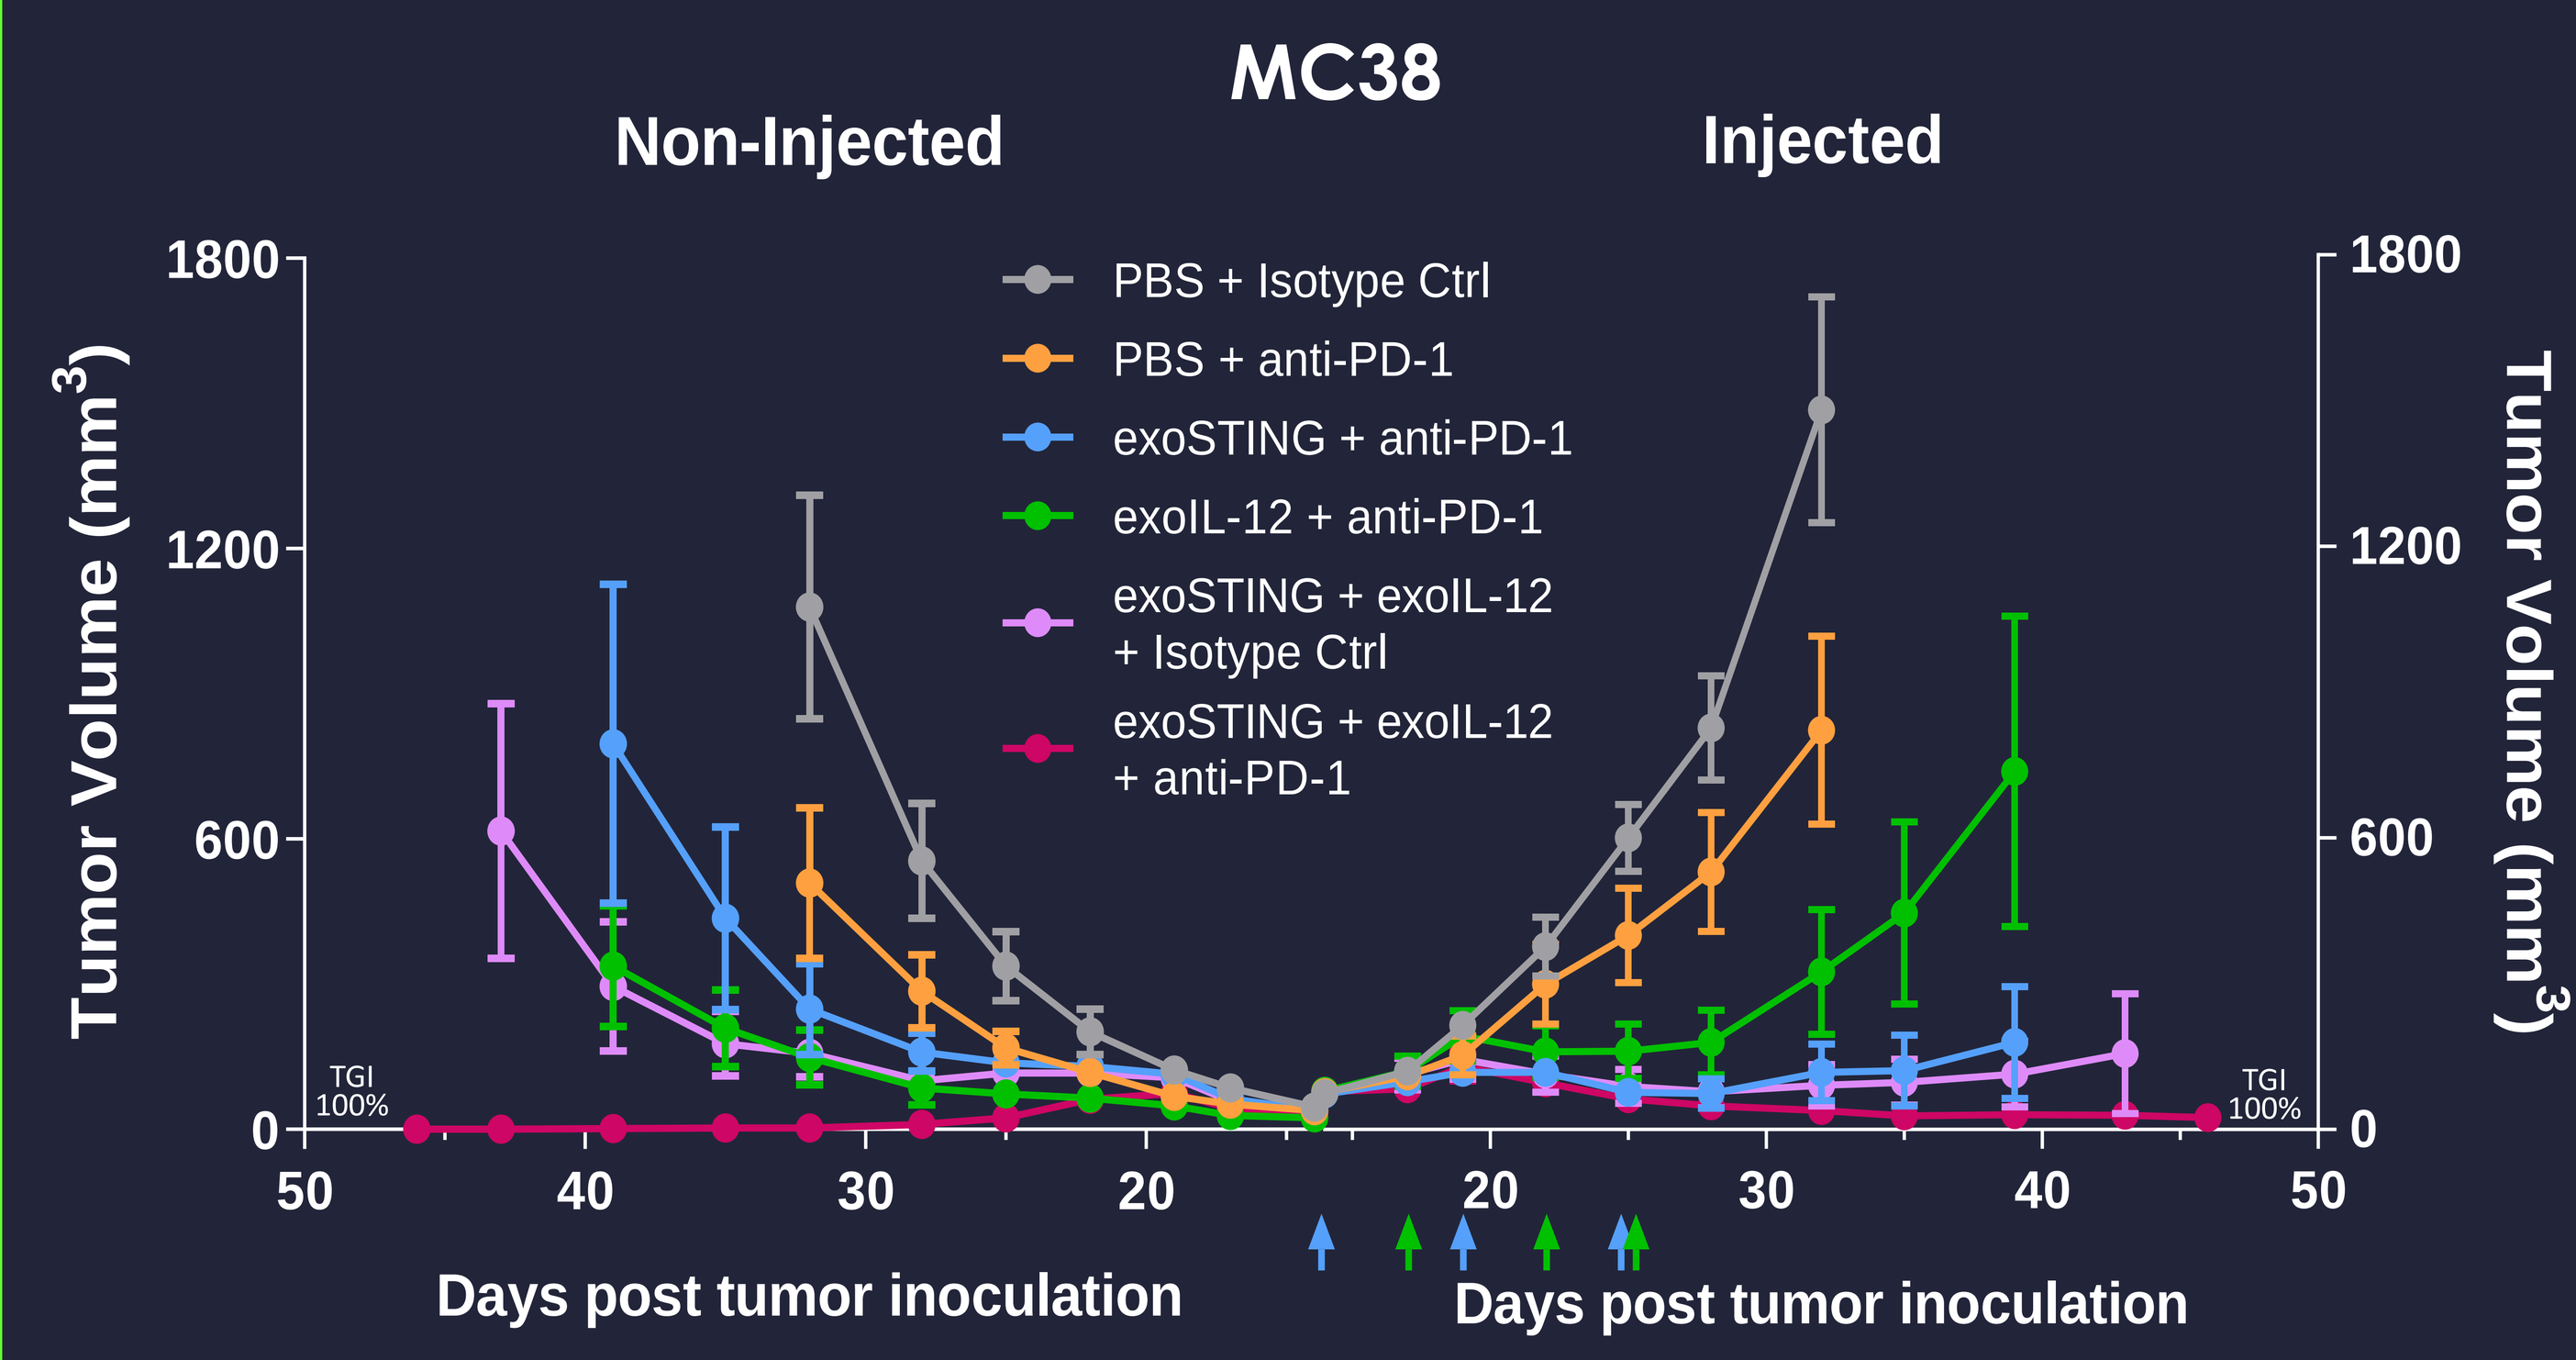
MC38
Non-Injected
Injected
PBS + Isotype Ctrl
PBS + anti-PD-1
exoSTING + anti-PD-1
exoIL-12 + anti-PD-1
exoSTING + exoIL-12 + Isotype Ctrl
exoSTING + exoIL-12 + anti-PD-1
TGI 100%
Tumor Volume (mm³)
Days post tumor inoculation

Triple combination therapy (n = 5) of exoSTING and exoIL12 reduced mean tumor volume in a dual flank MC38 subcutaneous mouse model resulting in 100% complete response rate. Dosing started 14 days post tumor inoculation for single agent exoSTING (q3d) or exoIL-12 (q2d). Combination therapy with the same dosing scheme with exoIL-12 was initiated two days post exoSTING as indicated by arrows. Mice also were dosed with anti-PD-1 or isotype control (IP).

5. exoSTING, exoIL-12, anti-PD-1 triple combination demonstrates improved survival and durable memory

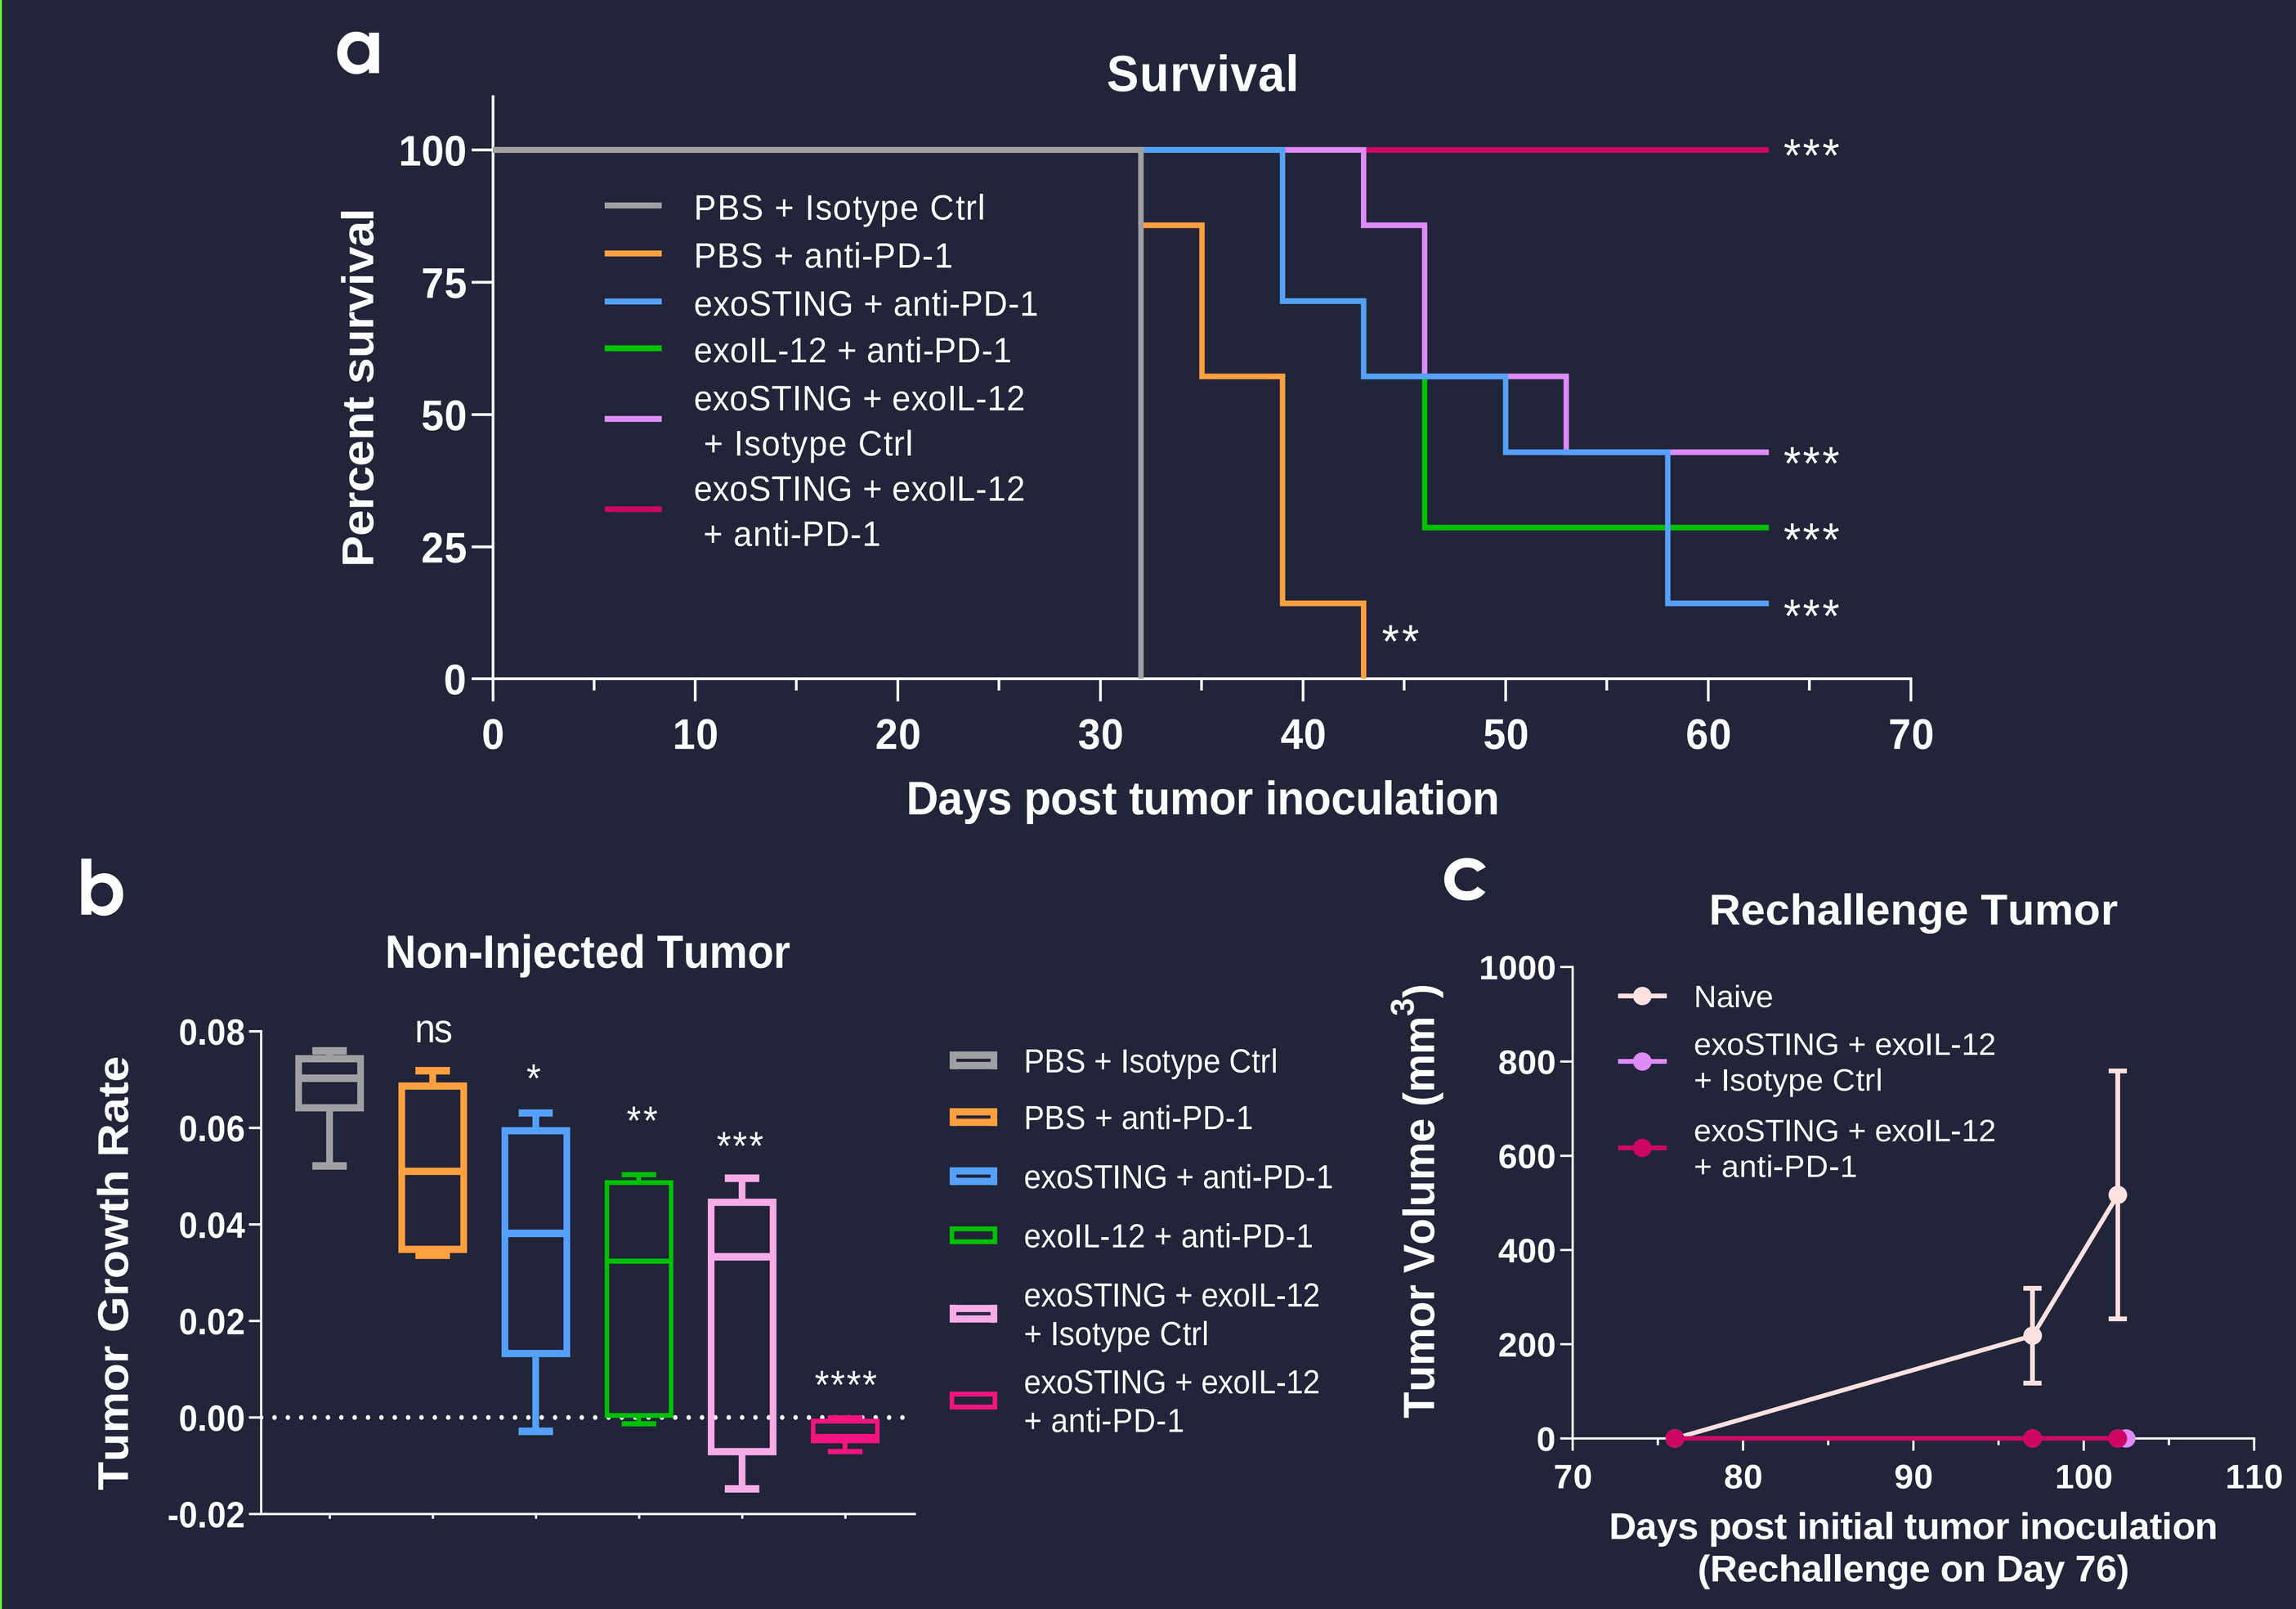
a
Survival
PBS + Isotype Ctrl
PBS + anti-PD-1
exoSTING + anti-PD-1
exoIL-12 + anti-PD-1
exoSTING + exoIL-12 + Isotype Ctrl
exoSTING + exoIL-12 + anti-PD-1
Percent survival
Days post tumor inoculation
b
Non-Injected Tumor
Tumor Growth Rate
c
Rechallenge Tumor
Naive
Tumor Volume (mm³)
Days post initial tumor inoculation
(Rechallenge on Day 76)

Triple intratumoral combination therapy (n = 5) of exoSTING and exoIL12. Dosing as indicated in previous panel. Mice were dosed with anti-PD-1 or isotype control (IP). Survival of triple combination (a) was 100% (*** $p=0.0003$, ** $p=0.0018$). Tumor growth rate of non-injected tumor demonstrated highly significant decrease (b) in tumor growth rate with exoSTING, exoIL-12, and triple anti-PD-1 combination (one-way ANOVA with Tukey's multiple comparison test; **** $p<0.0001$, *** $p=0.0006$, ** $p=0.0038$, * $p=0.0265$). No tumors grew following subcutaneous rechallenge with MC38 tumor cells in 100% responders 76 days post original tumor inoculation demonstrating lasting immunological memory (c).

6. Optimizing the dosing schedule for the triple combination therapy

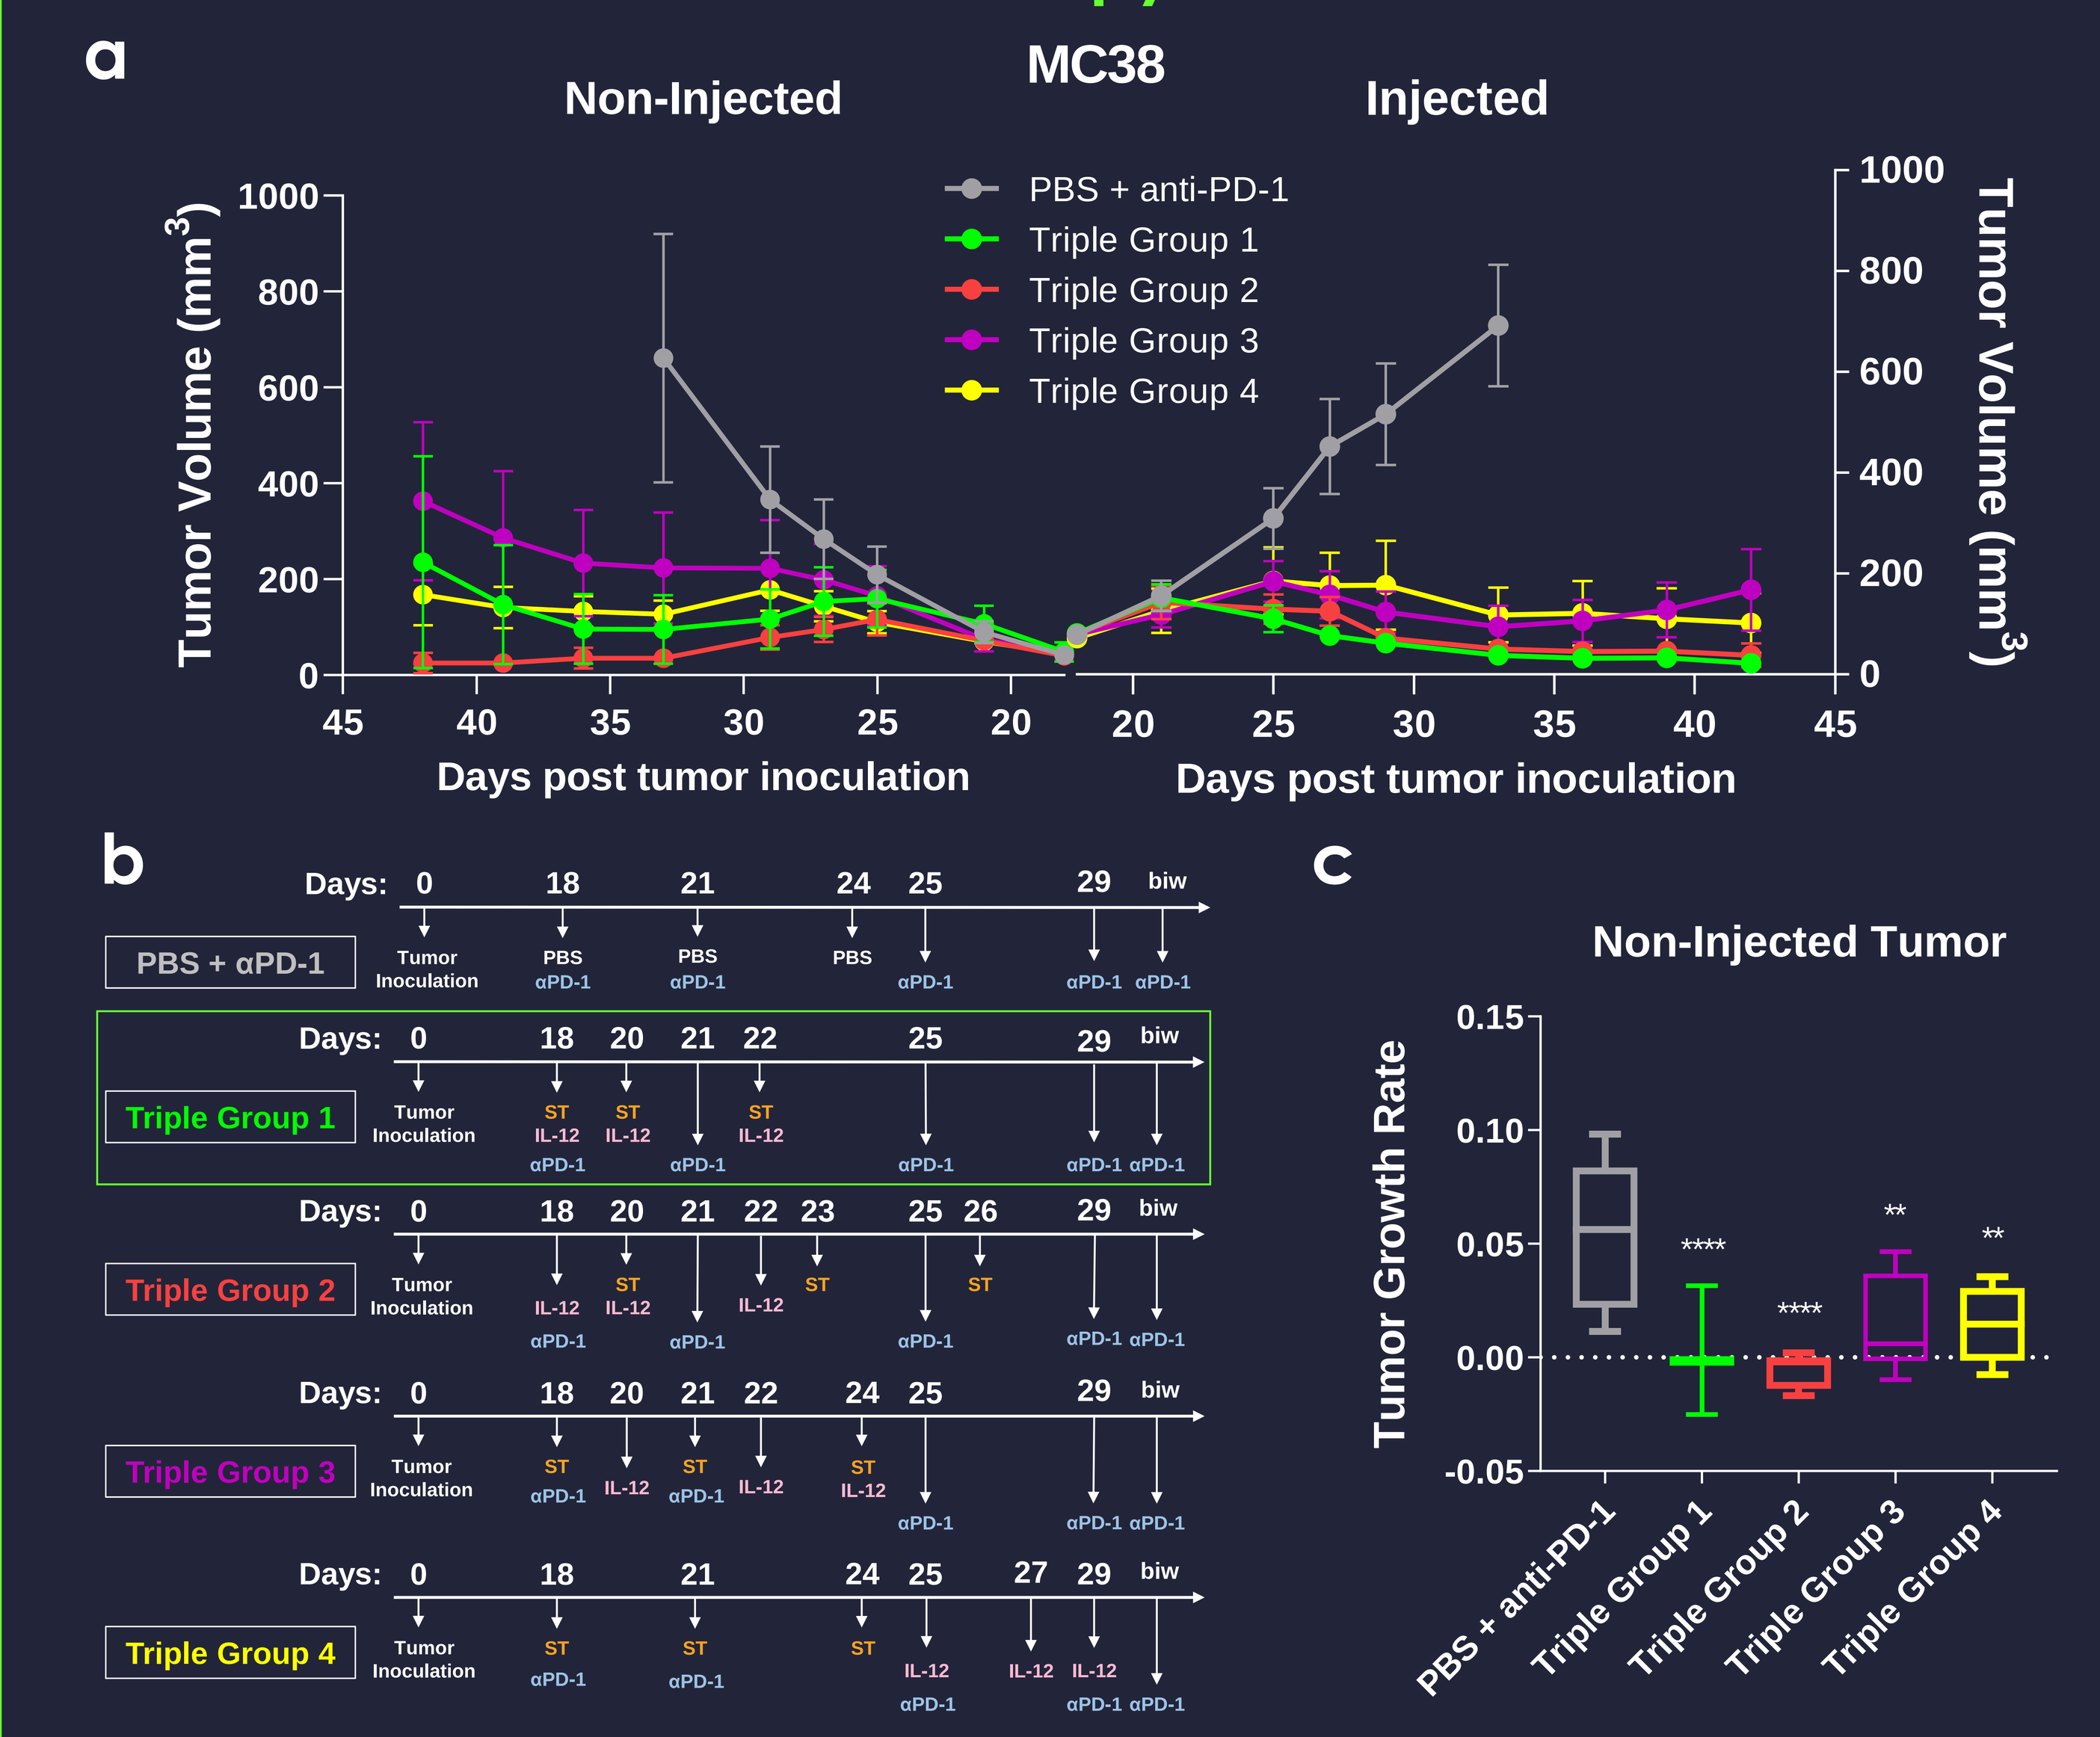
a
MC38
Non-Injected
Injected
PBS + anti-PD-1
Triple Group 1
Triple Group 2
Triple Group 3
Triple Group 4
Tumor Volume (mm³)
Days post tumor inoculation
b
PBS + αPD-1
Triple Group 1
Triple Group 2
Triple Group 3
Triple Group 4
Days:
Tumor Inoculation
c
Non-Injected Tumor
Tumor Growth Rate

Mean tumor volume in a dual flank MC38 subcutaneous mouse model (a). Mice were dosed intratumorally three times (n = 8), starting 18 days post tumor inoculation. Mice were also dosed intraperitoneally with anti-PD-1 or isotype control. All triple group combinations received exoSTING, exoIL-12, and anti-PD-1. Dosing scheme (b) of each group depicting days of intratumoral dosing. "ST" represents exoSTING, "IL-12" represents exoIL-12. Tumor growth rate of non-injected tumor demonstrates significant differences with all groups compared to PBS + anti-PD-1 with Triple Groups (c) 1 and 2 have lowest growth rate, validating choice of Group 1 for future studies (one-way ANOVA with Tukey's multiple comparison test; **** $p<0.0001$, ** $p=0.0042$ Triple Group 3, ** $p=0.0035$ Triple Group 4).

Summary

- exoSTING with anti-PD-1 Combination therapy significantly increases T cell infiltration into non-injected tumor.
- Intratumoral administration of exoSTING and exoIL-12 in the presence of systemically dosed anti-PD-1 significantly increased tumor inhibition, eliminating both injected and non-injected MC38 tumors.
- Triple combination demonstrated durable immunological memory in all complete responders with enhanced systemic anti-tumor immunity.
- exoSTING and exoIL-12 can be dosed simultaneously for optimal efficacy.
- exoSTING and exoIL-12 are both currently in clinical development.

Presented at the 36th Annual Meeting of the Society for Immuno-Therapy of Cancer, November 13th, 2021 in Washington, D.C. USA.