Generation Of Potent And High-Yield Exosome-Associated AAV (exoAAV) Using Engineered Exosomes

Russell McConnell, Bhargavee Gnanasambandam, Aaron Noyes, Sevda Lule, Palak Shah, Wei Zhang, Kevin Dooley, Tong Zi, Silvia Siso, Shil Patel, Jonathan D. Finn, Sriram Sathyanarayanan
Codiak BioSciences, Cambridge, MA

BACKGROUND

Adeno-associated virus (AAV) is a commonly used gene therapy vector due to its excellent safety profile, stability, and duration of transgene expression. One major limitation of current AAV vectors is neutralizing antibodies (nAb), which are present in many potential patients (preventing treatment) and develop in all patients following an initial dose (preventing re-administration). Encapsulation within exosomes (exoAAV) is a potential strategy to protect AAV from antibody-mediated neutralization, allowing efficient transduction even in the presence of high nAb titers. We have developed a method for loading AAV into exosomes using camelid nanobodies. Engineered exoAAV yields are multi-log higher than endogenously loaded exoAAV while still retaining desirable characteristics of exoAAV.

SUMMARY

- exoAAV resists neutralizing antibodies *in vitro*
- exoAAV and AAV show different cellular tropism *in vivo*
- Yields of exoAAV from unmodified producer cells are low, and most secreted AAV is not exosome-associated
- exoAAV yield can be increased using engineered exosomes
- Engineered exoAAV retains transduction ability

CONTACT

Russell McConnell, PhD
Codiak BioSciences
Email: russell.mcconnell@codiakbio.com
Website: www.codiakbio.com

Some AAV is naturally encapsulated within exosomes (exoAAV)

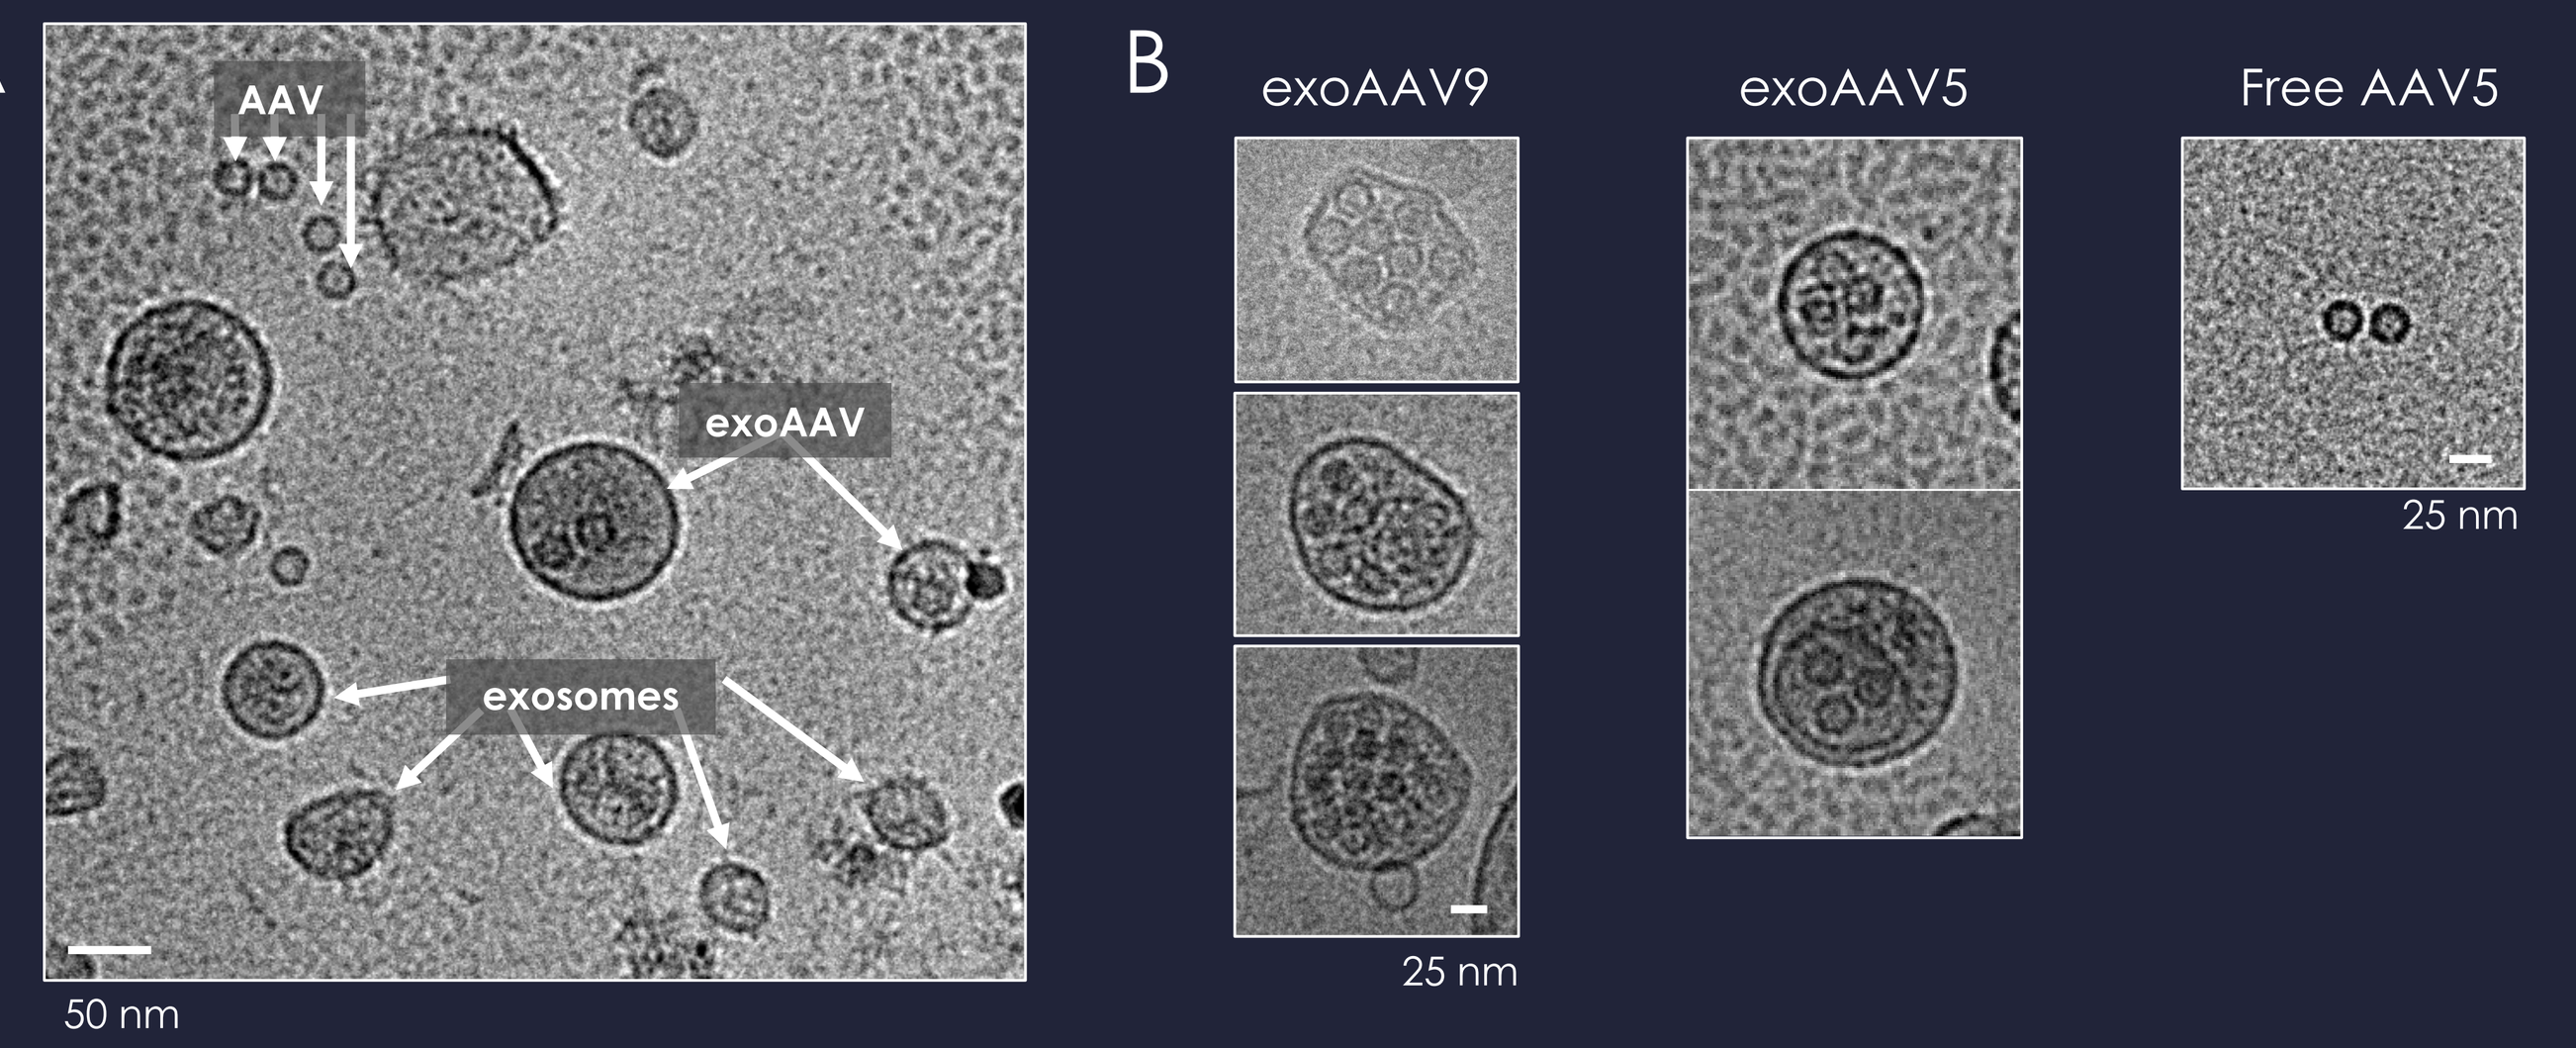
A
AAV
exoAAV
exosomes
50 nm
B
exoAAV9
exoAAV5
Free AAV5
25 nm

Figure 1. Ultrastructural analysis of exoAAV produced from transiently transfected cells.
A) Cryo-EM image showing that purified exoAAV contains a mix of exosomes, free AAV, and exoAAV.
B) exoAAV can be made with different AAV serotypes.

Very little secreted AAV is naturally exosome-associated

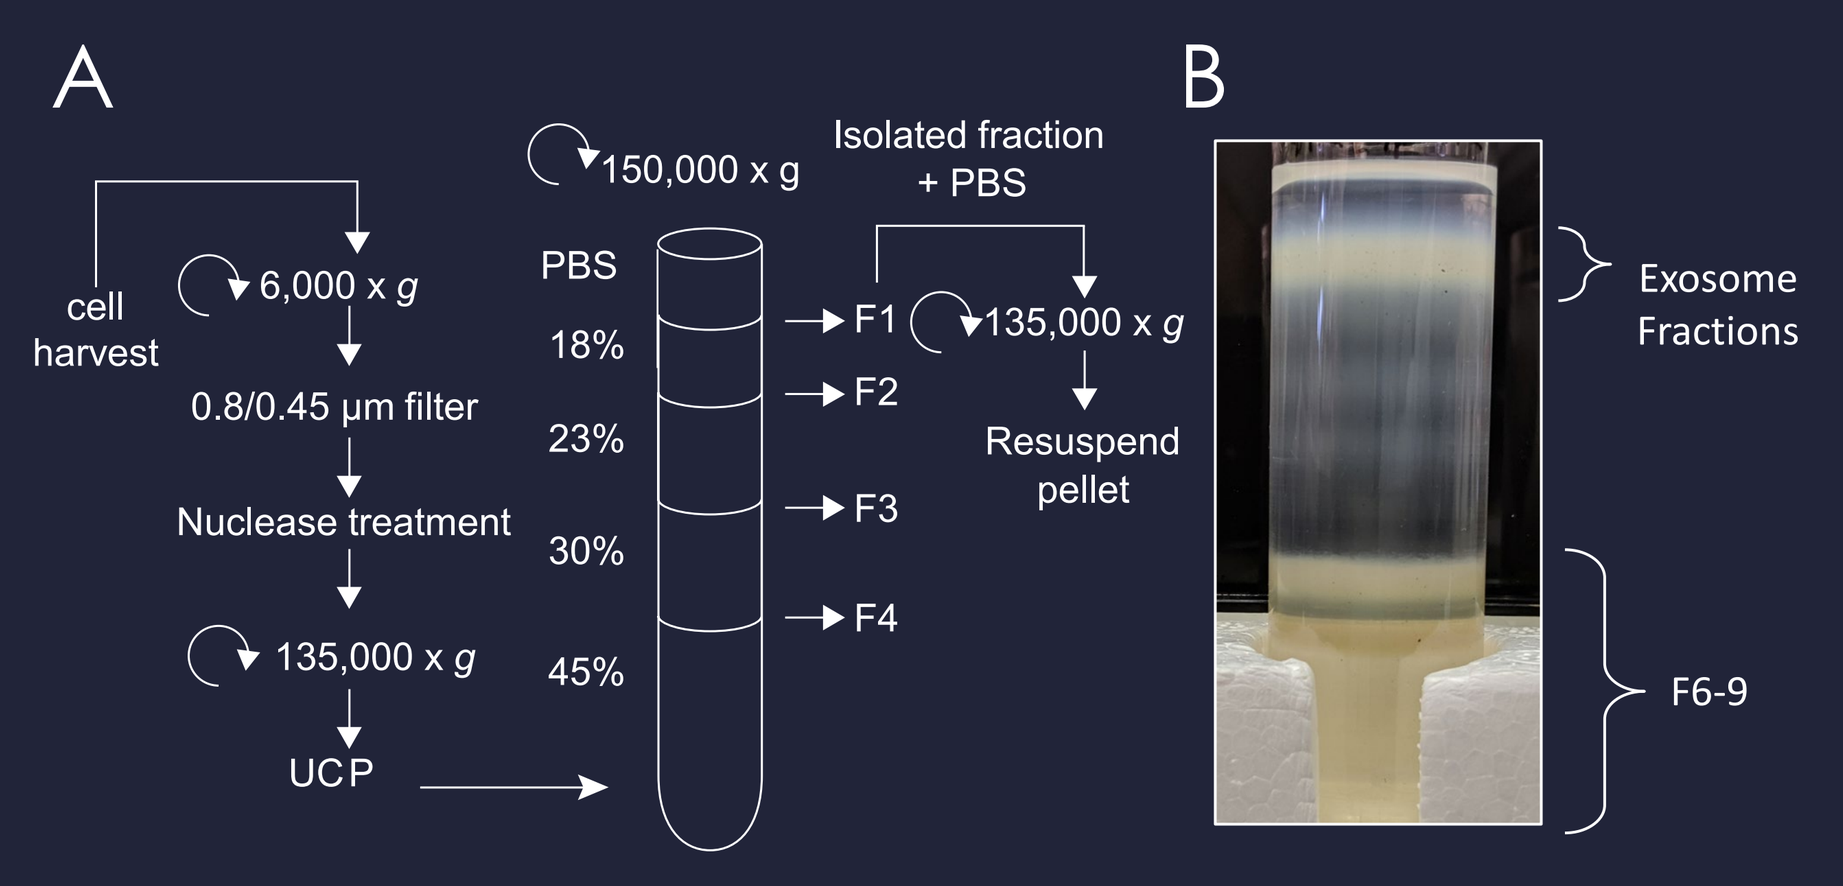
A
cell harvest
6,000 x g
0.8/0.45 µm filter
Nuclease treatment
135,000 x g
UCP
150,000 x g
PBS
18%
23%
30%
45%
F1
F2
F3
F4
Isolated fraction + PBS
135,000 x g
Resuspend pellet
B
Exosome Fractions
F6-9

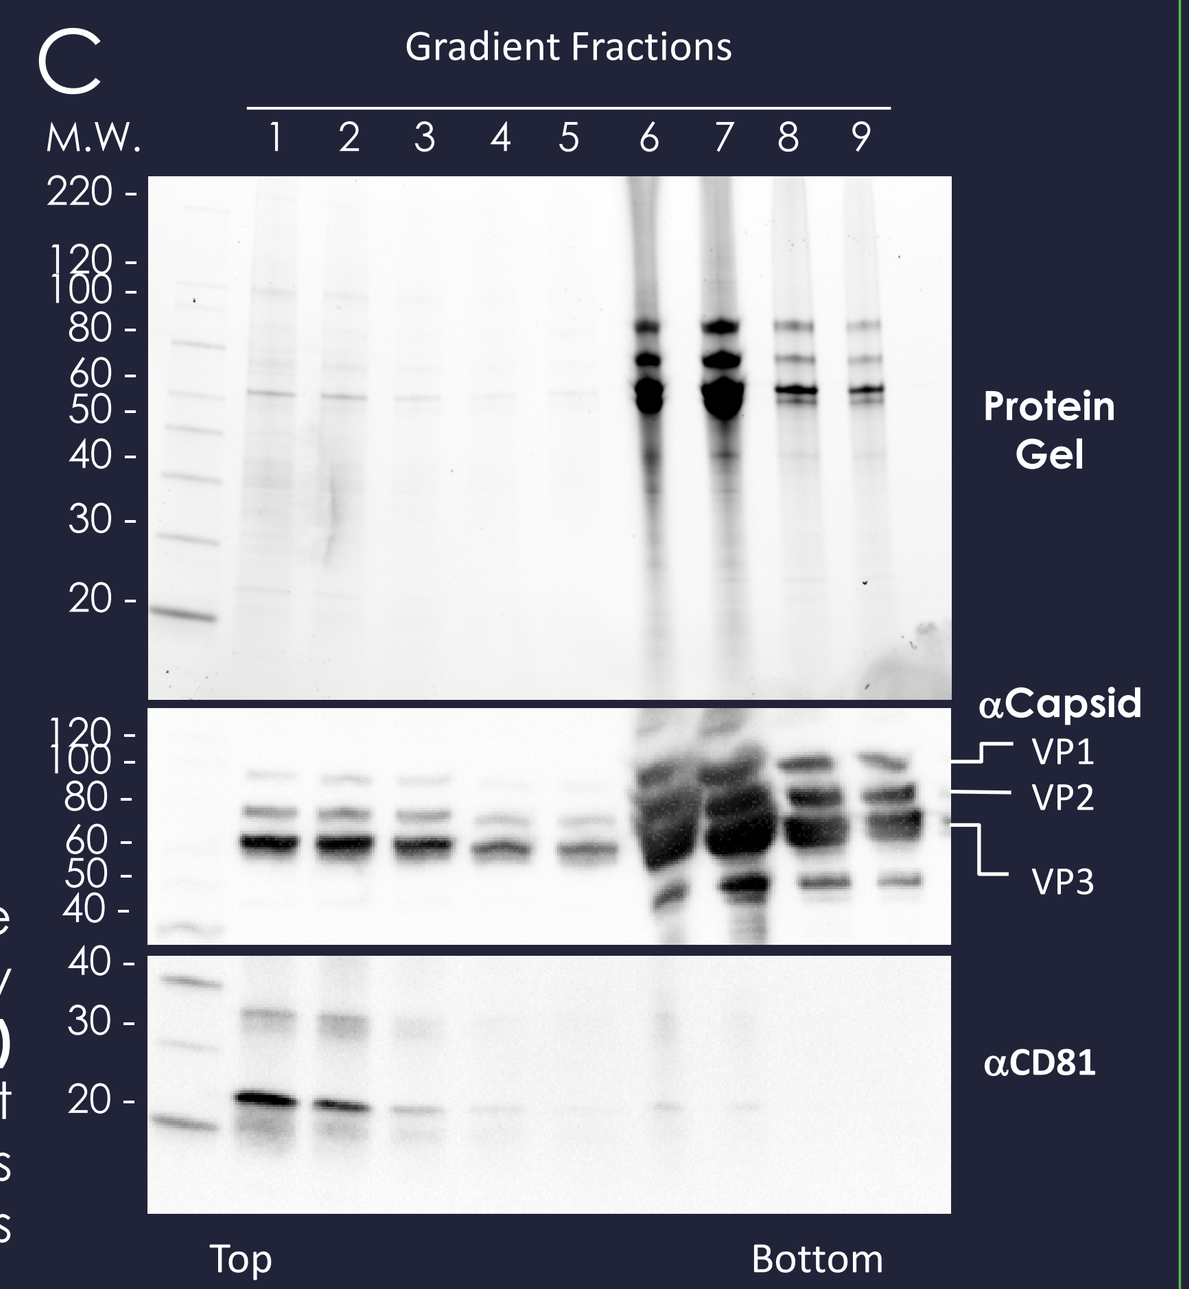
C
Gradient Fractions
M.W.
1 2 3 4 5 6 7 8 9
Protein Gel
αCapsid
VP1
VP2
VP3
αCD81
Top
Bottom

Figure 2. exoAAV purification. A) Schematic of purification procedure used to purify exoAAV. A combination of differential and density gradient centrifugation was used to separate exoAAV from free AAV. **B)** Image of Iodixanol density gradient. **C)** SDS-PAGE and Western blot analysis of density gradient fractions. Exosome-containing fractions (CD81+, F1-3) also contain AAV. The majority of AAV is found in fractions depleted of exosomes (F6-9).

exoAAV resists neutralizing antibodies *in vitro*

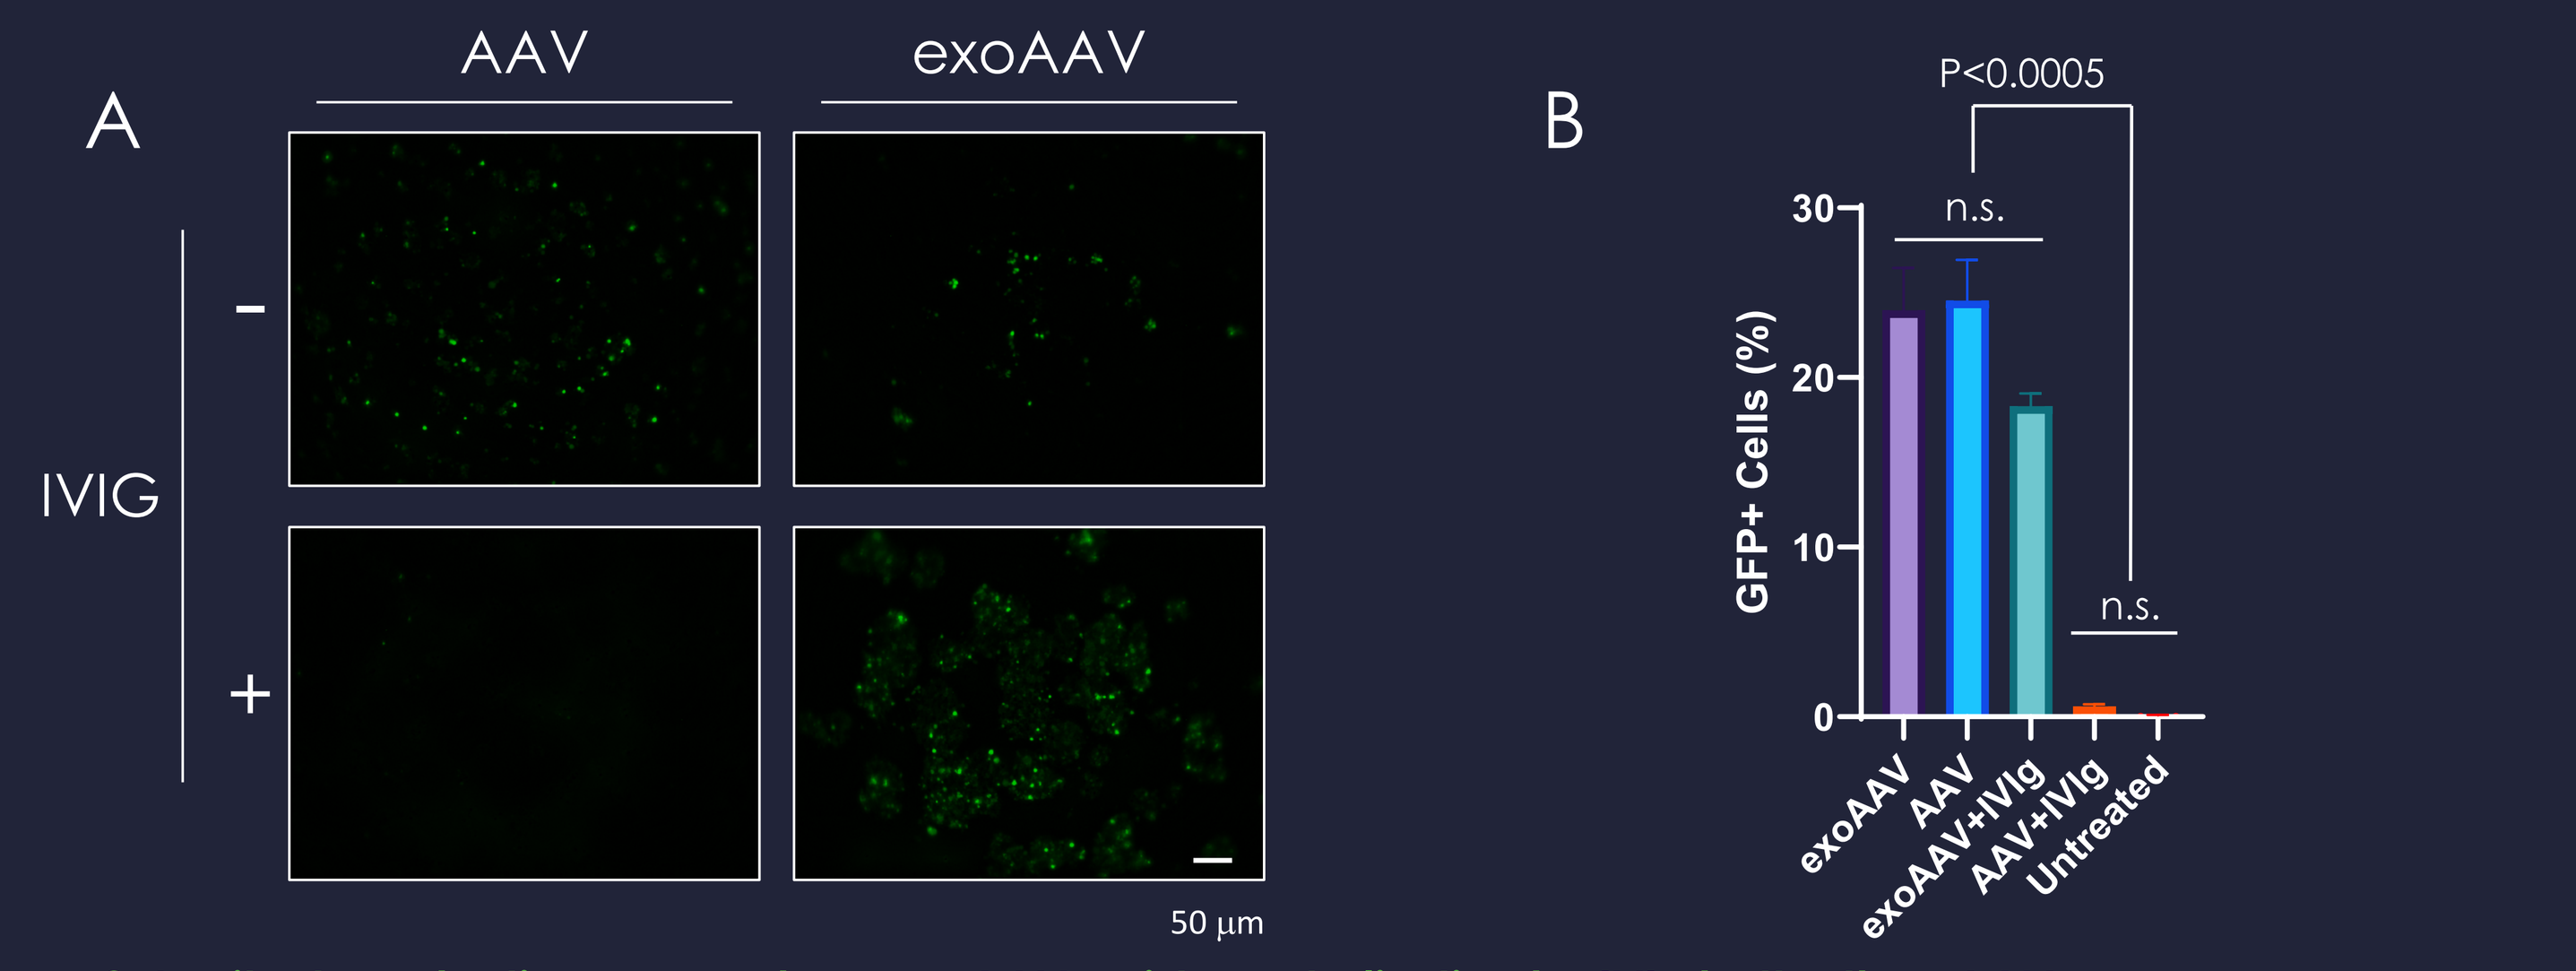
A
AAV
exoAAV
IVIG
-
+
50 µm
B
P<0.0005
n.s.
n.s.
GFP+ Cells (%)
exoAAV
AAV
exoAAV+IVIg
AAV+IVIg
Untreated

Figure 3. *In vitro* transduction assays show exoAAV resists neutralization by IVIg better than AAV.
A) Representative fluorescence images of HEK cells showing GFP expression 3 days post-treatment with AAV or exoAAV in the absence or presence of 100 µg/mL human IVIg. **B)** GFP expression analyzed by flow cytometry.

ExoAAV shows different cellular tropism than AAV *in vivo*

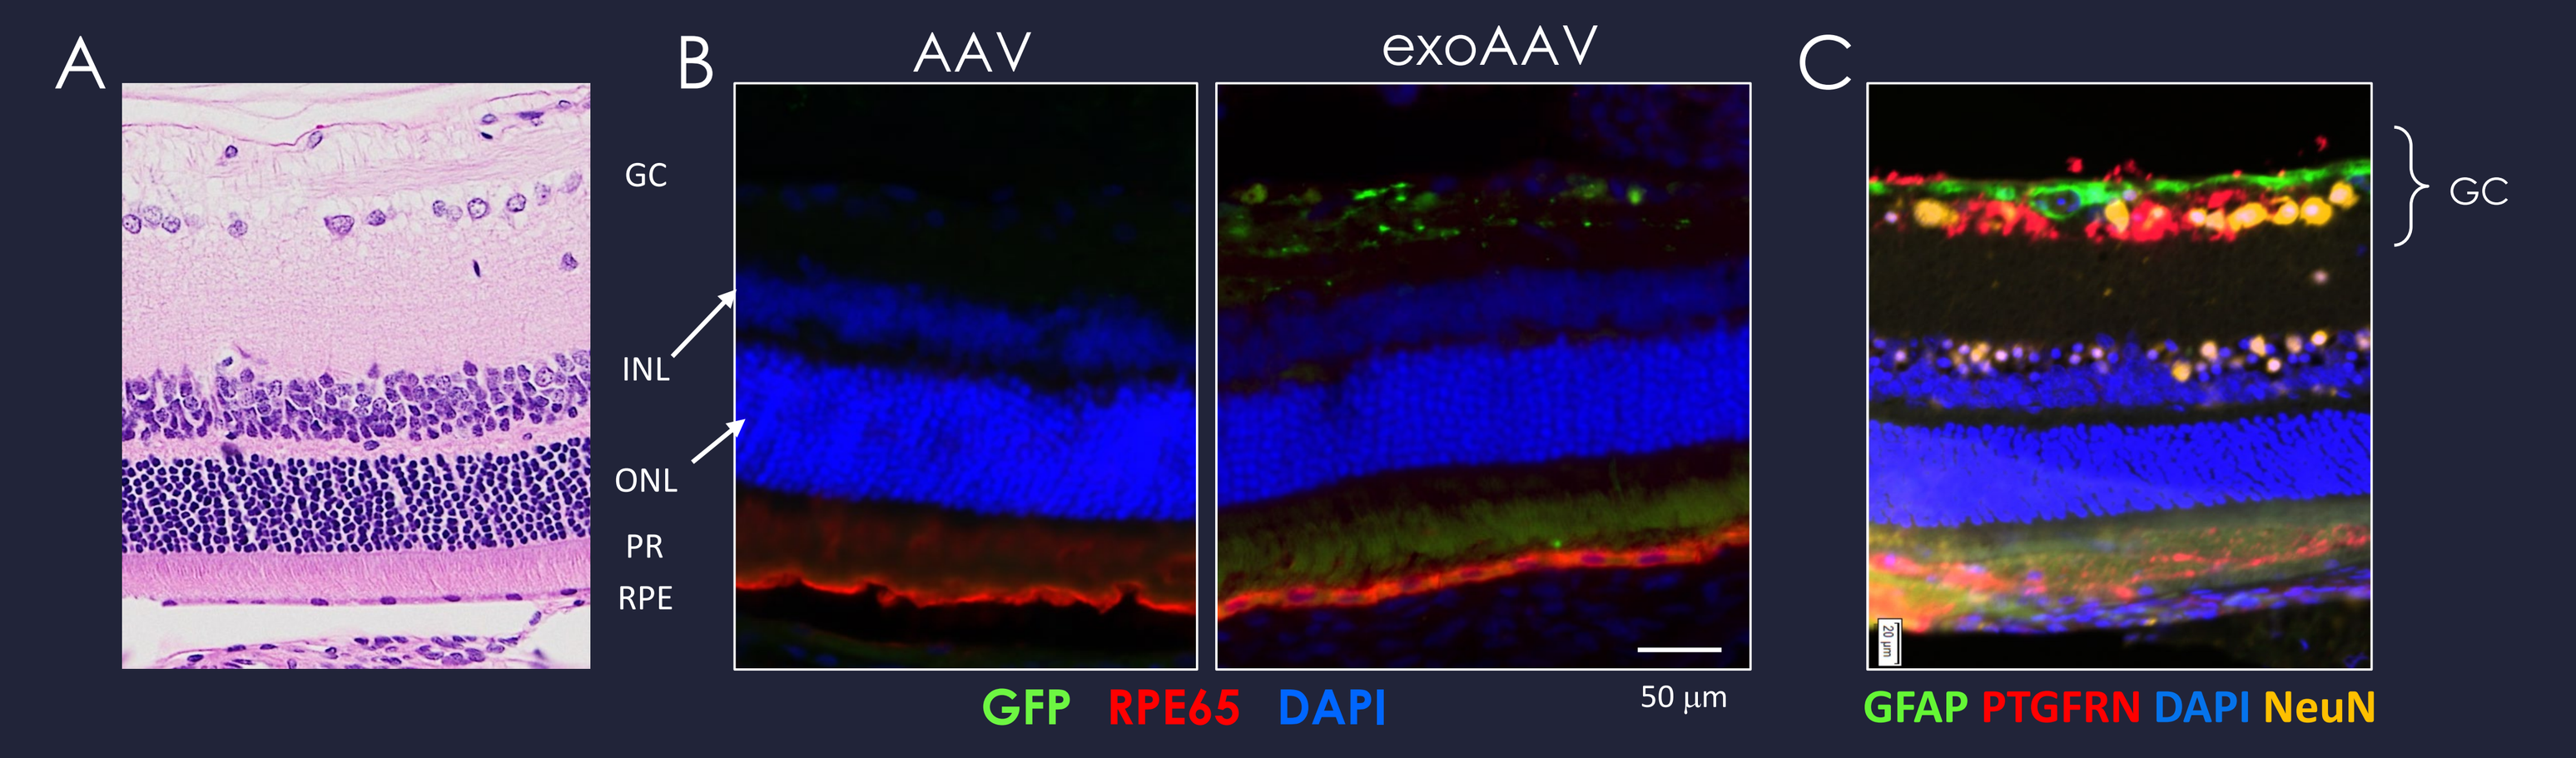
A
GC
INL
ONL
PR
RPE
B
AAV
exoAAV
GFP RPE65 DAPI
50 µm
C
GC
GFAP PTGFRN DAPI NeuN

Figure 4. GFP transduction by intravitreal injection of exoAAV mirrors exosome biodistribution in rats.
A) H&E-stained paraffin section showing retinal morphology. GC- ganglion cells, INL-inner nuclear layer, ONL-outer nuclear layer, PR-photoreceptors, RPE-retinal pigment epithelium. **B)** Immunohistochemistry showing GFP expression 21 days after intravitreal injection of AAV or exoAAV carrying GFP transgene (1e10 GC per eye). Distal to the injection site, GFP expression was observed in the GC layer in exoAAV-injected eyes, but not in AAV-injected eyes. **C)** Exosome biodistribution assessed 4 hours following intravitreal injection. Staining to detect an exosome marker (human PTGFRN, red) shows exosome localization in the GC layer.

Exosome engineering strategy

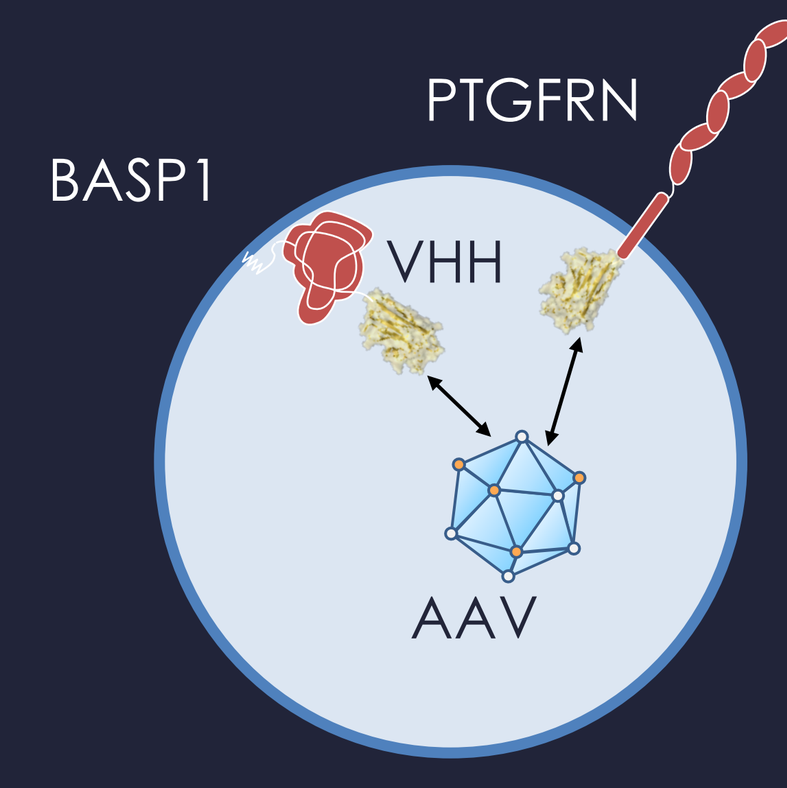
BASP1
PTGFRN
VHH
AAV

Design characteristics for anti-AAV affinity ligand to increase exoAAV yield

- Must be able to bind AAV intracellularly
- Amenable to protein engineering
- Increase loading without inhibiting transduction
- Ease of screen to identify candidate binders

Screen to identify novel AAV-binding affinity agents

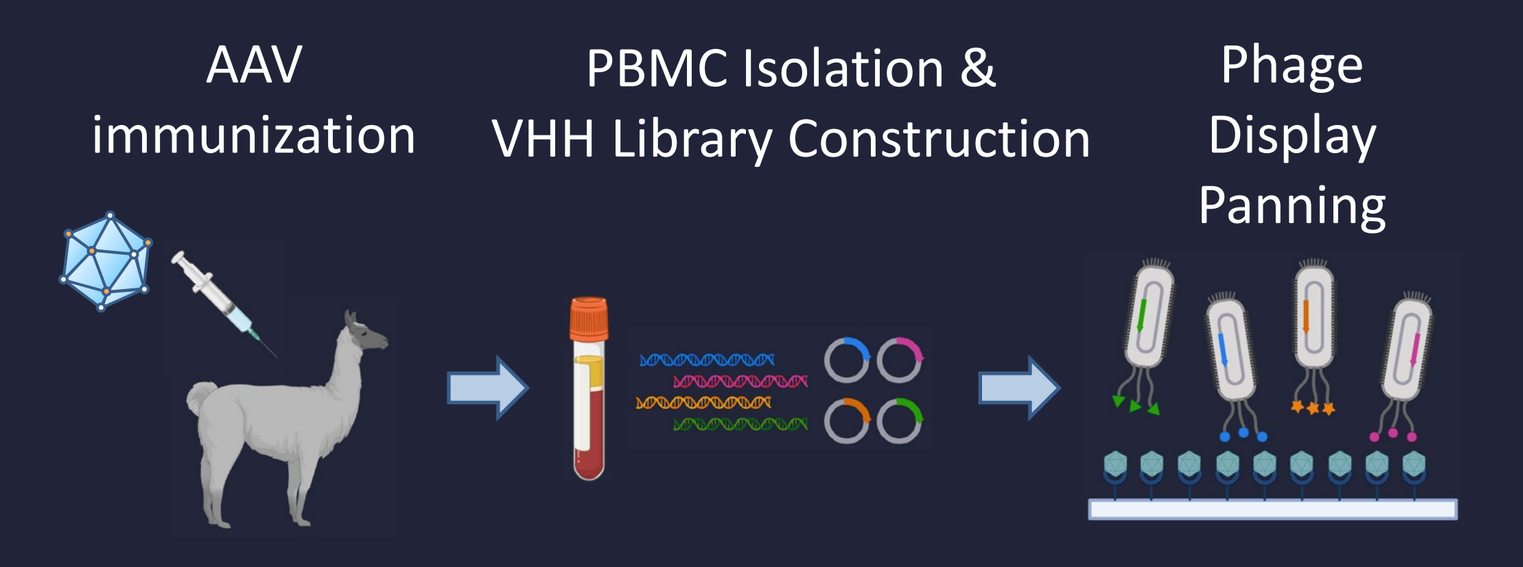
AAV immunization
PBMC Isolation & VHH Library Construction
Phage Display Panning

Figure 5. Identification of novel camelid nanobody sequences. Alpaca was immunized with AAV, and blood tested by ELISA for the presence of AAV-binding antibodies. Once acceptable titers were achieved, PBMCs were isolated, RNA extracted, and a cDNA library constructed. Plasmid library was panned using bacteriophage display to identify VHH sequences.

Some VHH show favorable low activity in neutralization assays

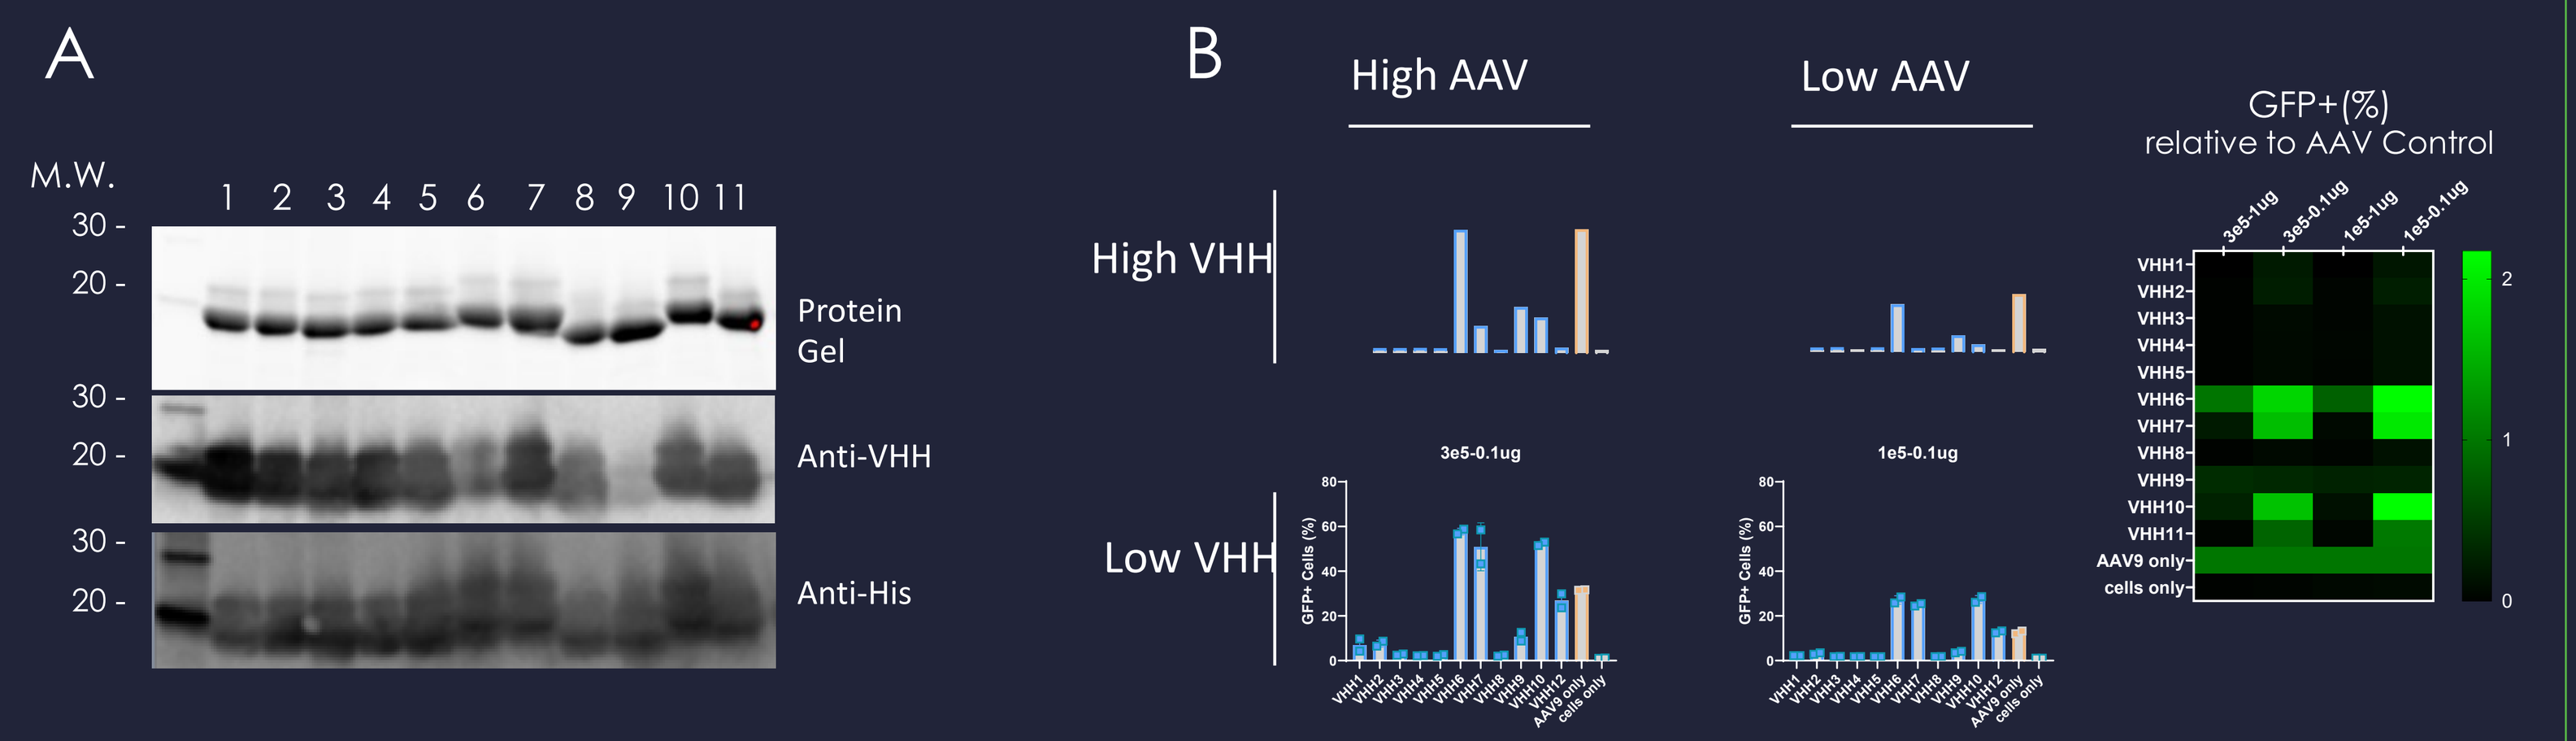
A
M.W.
1 2 3 4 5 6 7 8 9 10 11
Protein Gel
Anti-VHH
Anti-His
B
High AAV
Low AAV
High VHH
Low VHH
GFP+(%) relative to AAV Control

Figure 6. *In vitro* neutralization assay with recombinant nanobodies. A) His-tagged Nanobodies were expressed in Expi293 cells and purified using IMAC and SEC chromatography. All constructs showed the expected molecular weight of ~15 kDa and were detected by an anti-camelid VHH antibody. **B)** We sought to determine which VHH constructs would not neutralize AAV transduction by adding AAV carrying a GFP transgene to HeLa cells +/- recombinant VHH at the indicated concentrations.

Exosome engineering increases AAV loading

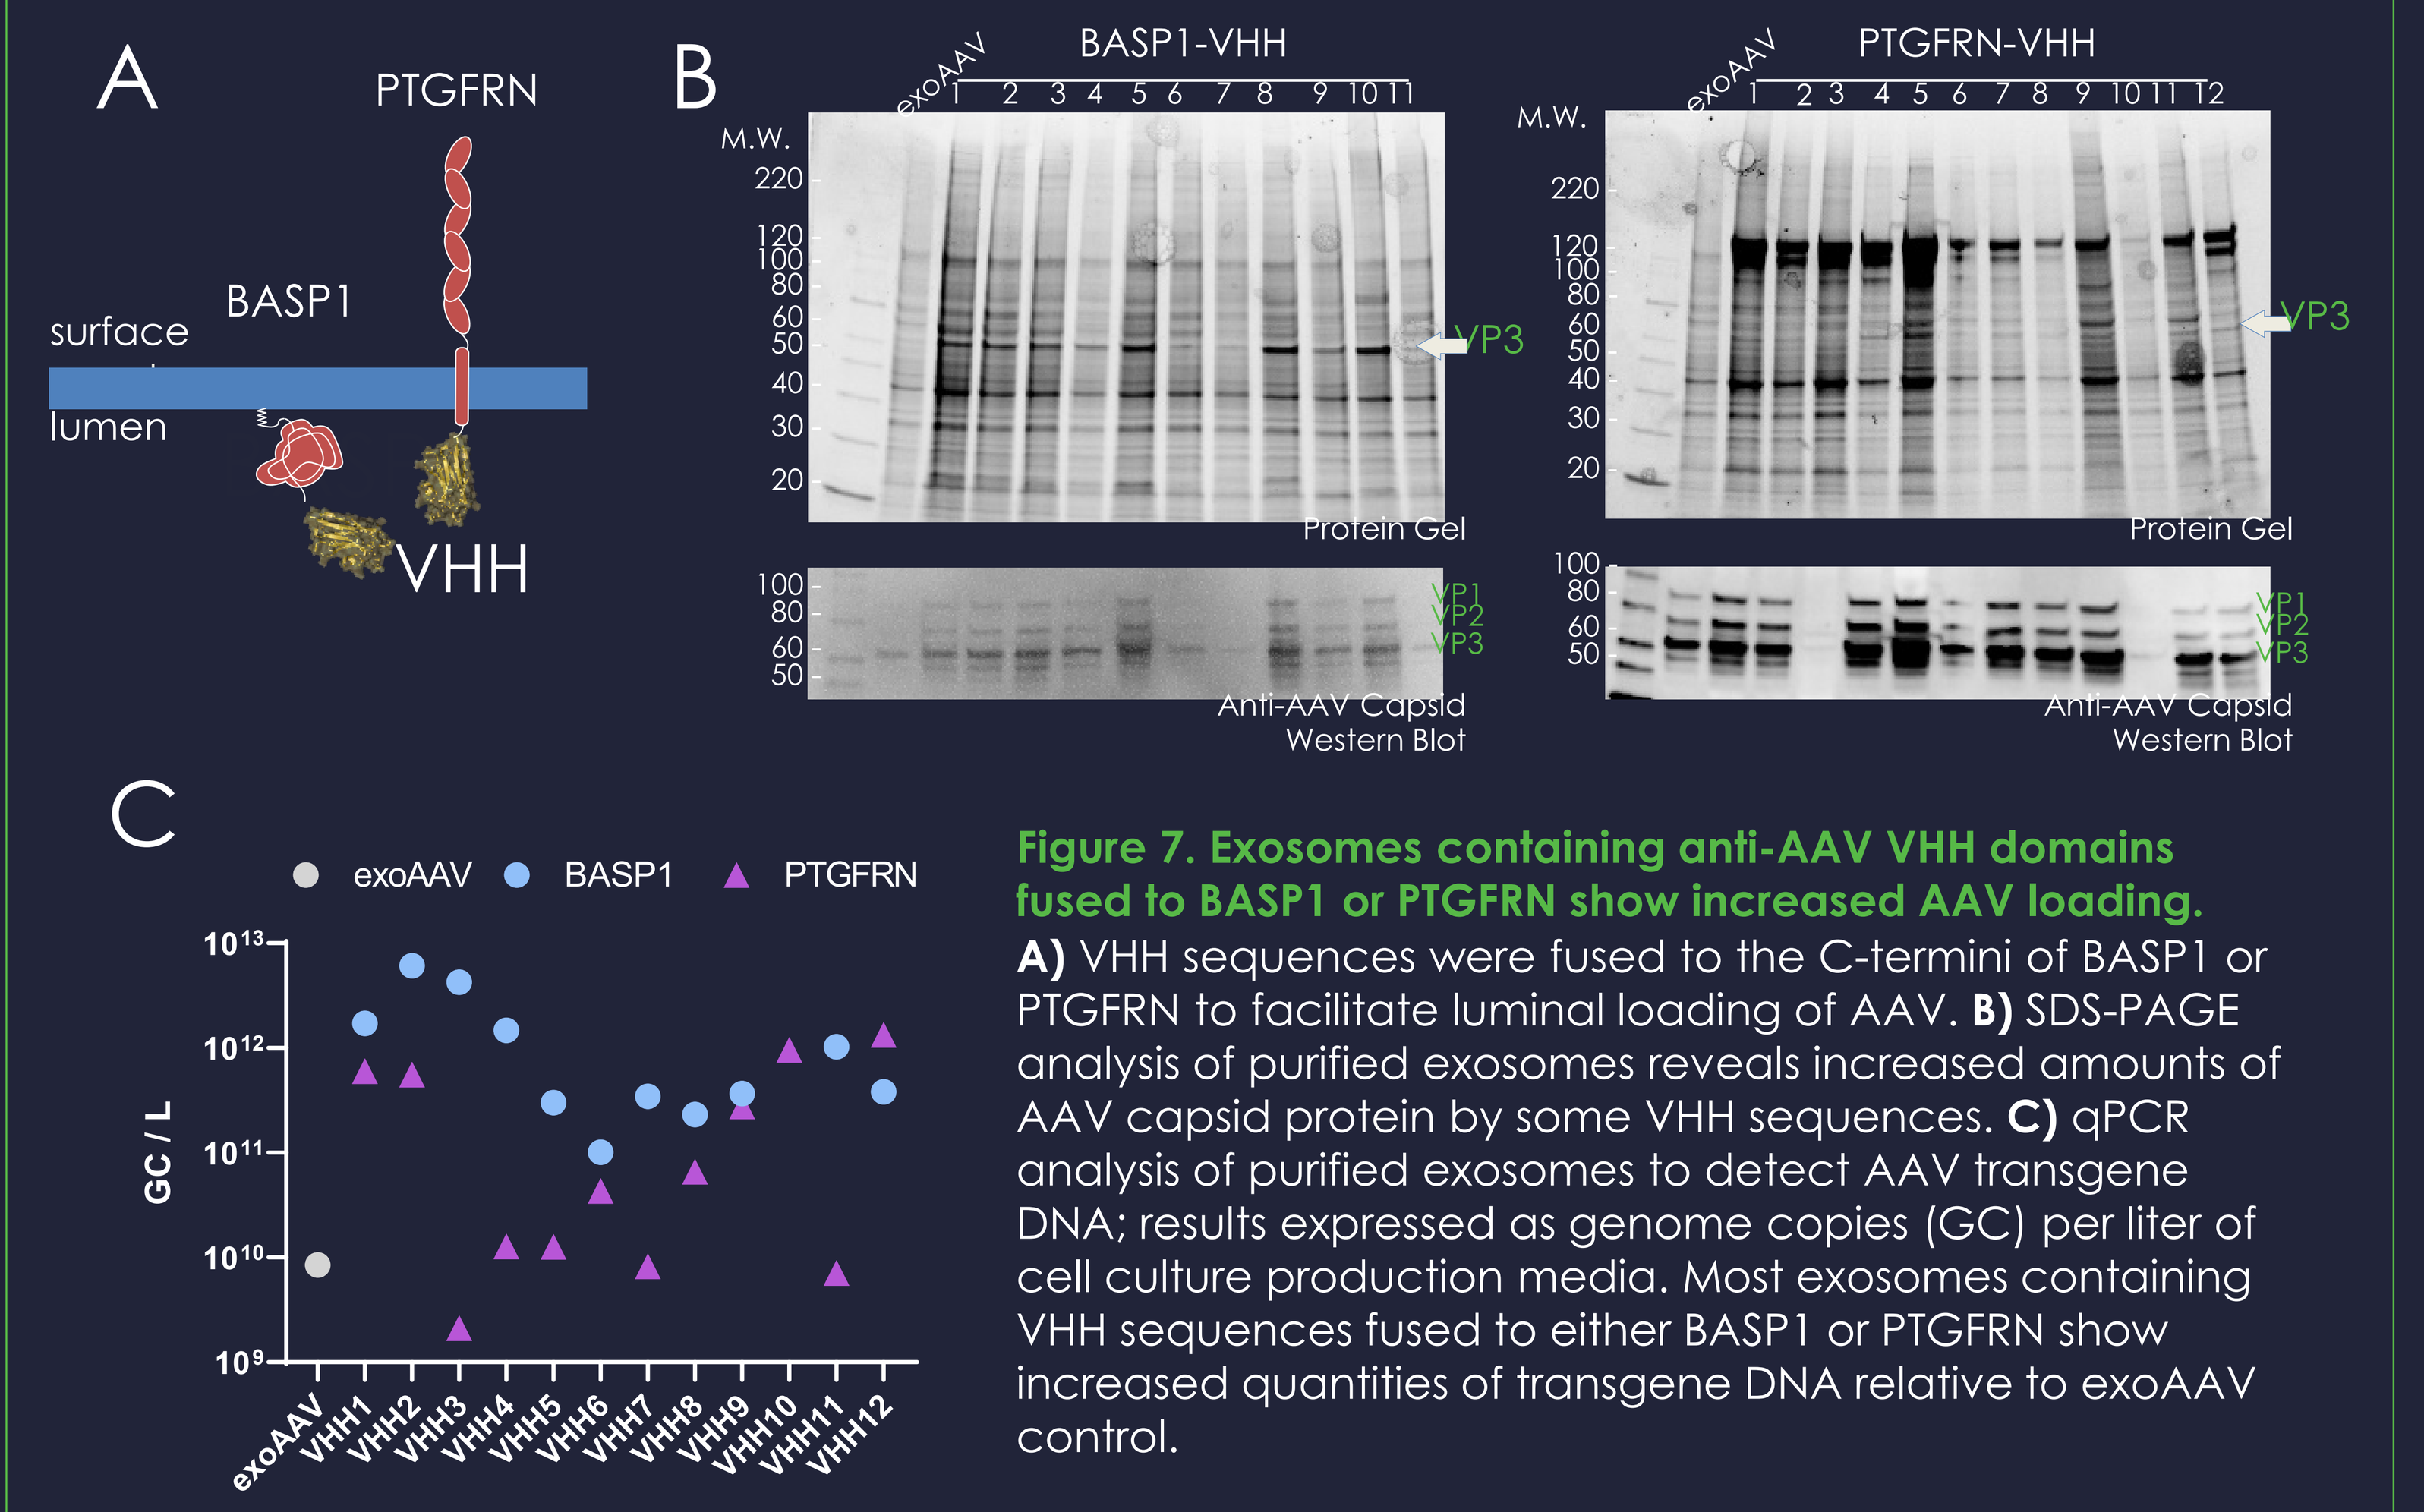
A
PTGFRN
BASP1
surface
lumen
VHH
B
BASP1-VHH
PTGFRN-VHH
exoAAV
M.W.
VP3
Protein Gel
VP1
VP2
VP3
Anti-AAV Capsid Western Blot
C
exoAAV
BASP1
PTGFRN
GC / L

Figure 7. Exosomes containing anti-AAV VHH domains fused to BASP1 or PTGFRN show increased AAV loading.
A) VHH sequences were fused to the C-termini of BASP1 or PTGFRN to facilitate luminal loading of AAV. **B)** SDS-PAGE analysis of purified exosomes reveals increased amounts of AAV capsid protein by some VHH sequences. **C)** qPCR analysis of purified exosomes to detect AAV transgene DNA; results expressed as genome copies (GC) per liter of cell culture production media. Most exosomes containing VHH sequences fused to either BASP1 or PTGFRN show increased quantities of transgene DNA relative to exoAAV control.

Engineered exoAAV transduces cells *in vitro*

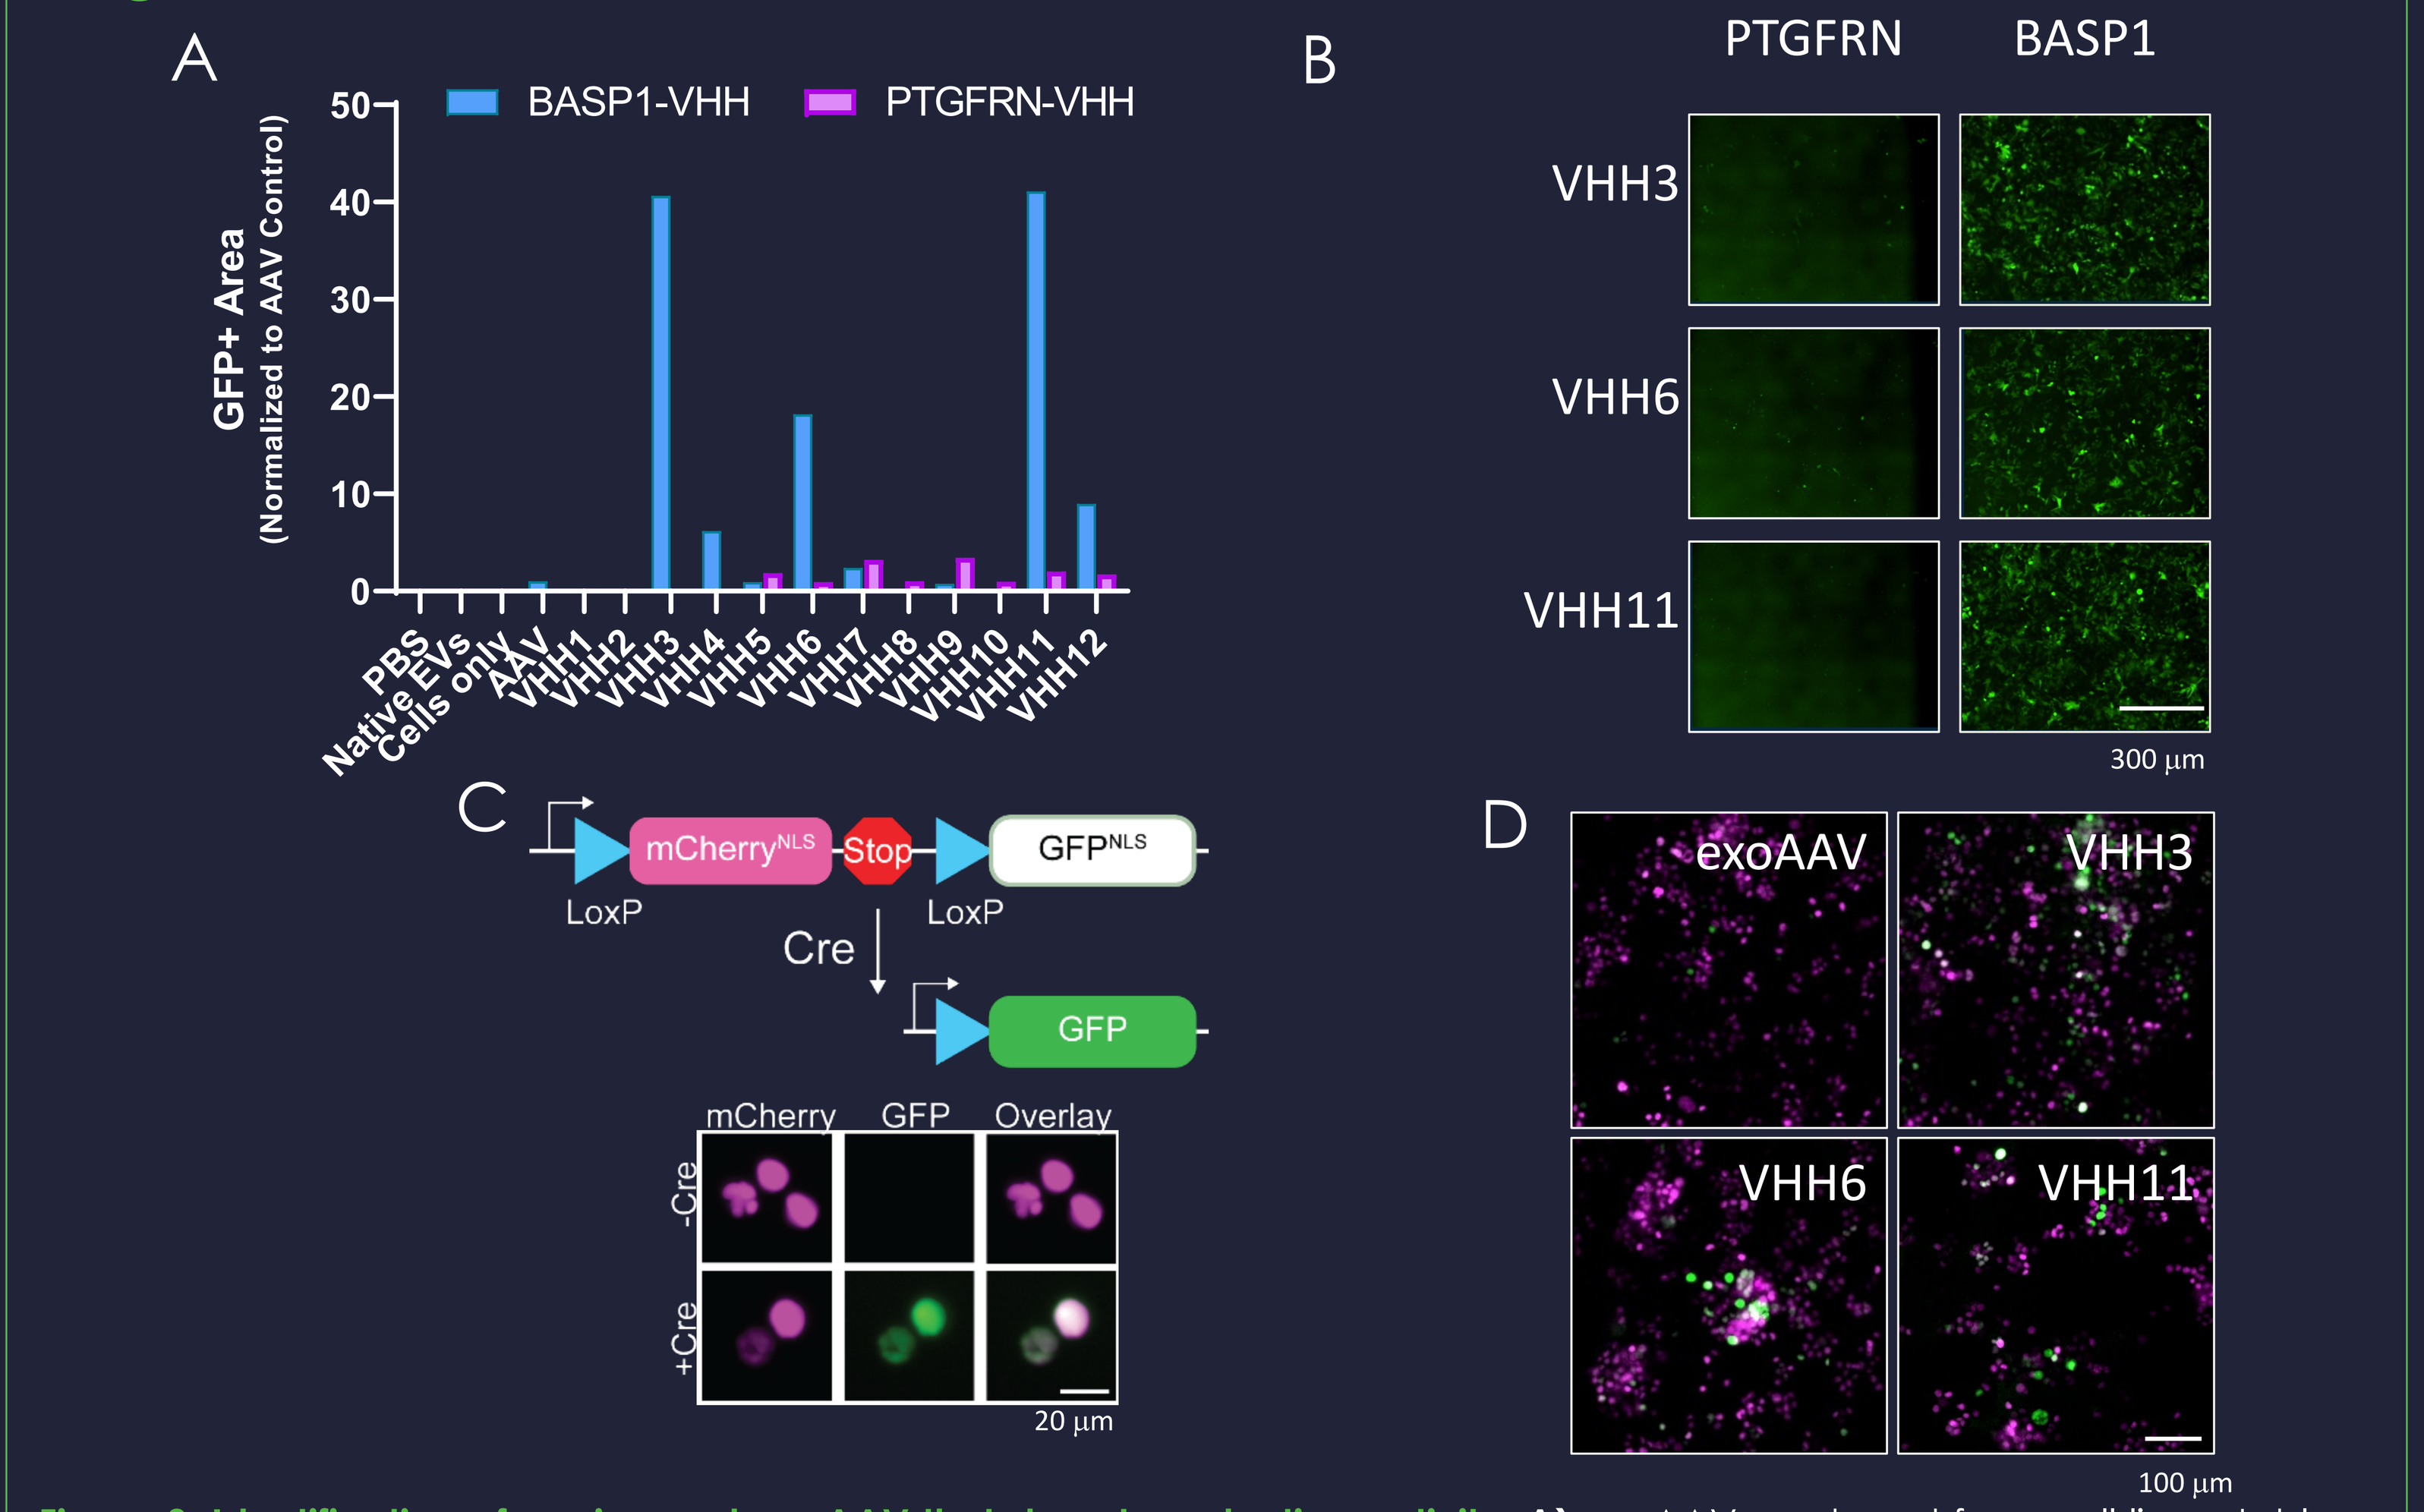
A
BASP1-VHH
PTGFRN-VHH
GFP+ Area (Normalized to AAV Control)
B
PTGFRN
BASP1
VHH3
VHH6
VHH11
300 µm
C
mCherry
Stop
GFP
LoxP
Cre
mCherry
GFP
Overlay
-Cre
+Cre
20 µm
D
exoAAV
VHH3
VHH6
VHH11
100 µm

Figure 8. Identification of engineered exoAAV that show transduction activity. A) exoAAV produced from cell lines stably expressing the indicated constructs were added to HeLa cells and GFP fluorescence quantitated 4 days later. **B)** Select images from the *in vitro* assay presented in panel A. VHH sequences 3, 6, and 11 showed significant transduction relative to other sequences and to AAV controls. Notably, BASP1 fusions of these VHH sequences showed significantly higher activity than the corresponding PTGFRN fusions. **C)** Cre "stoplight" red-to-green reporter system in HEK293 cells. **D)** Selected VHH-expressing cell lines were used to generate exoAAV carrying a Cre transgene. All three of the VHH sequences showed significant transduction when added to Cre reporter cells compared to an un-engineered exoAAV control.